A Mixed-Methods Study of Rapid ART in People Living with Advanced HIV in the Rio Grande Valley

Linda Mercado,[1,2] Elias Cantu,[1,2] Seth Francis-Graham,[3] Cameron Costello,[3] Eve Sullivan,[3] Hannah Borda,[4] Brenda Ng,[5] Sha McCoy[6]

[1]Valley AIDS Council, Harlingen, TX, USA; [2]Valley AIDS Council, McAllen, TX, USA; [3]Costello Medical, London, UK; [4]Costello Medical, Boston, MA, USA; [5]Gilead Sciences, Foster City, CA, USA; [6]Gilead Sciences, Houston, TX, USA

Study Design Objectives

- **AHORA was a mixed-methods study. The quantitative component was a single-arm, open label, ambidirectional, observational case series, which evaluated time to viral suppression and CD4 recovery/immune reconstitution in people with advanced HIV disease (AHD) rapidly initiating bictegravir/emtricitabine/tenofovir alafenamide (B/F/TAF) between January 2021–June 2023 at two clinics in the Rio Grande Valley (RGV).**
- **The qualitative component was a nested evaluation exploring reasons for delayed diagnosis, challenges with accessing care, and aspects of the rapid start treatment program that facilitated engagement. Quantitative and qualitative data were triangulated in a convergent parallel study design.[1]**

Background

- In the RGV in Texas, 93% of the population identifies as Latine and one-fourth live below the federal poverty line.[2]
- In 2021, between 63.6 and 218.4 people per 100,000 were living with HIV in the RGV,[3] many of whom face socioeconomic disparities, presenting barriers to diagnosis that increase the risk of AHD.[4]
- Rapid initiation of ART is now considered best practice, particularly for AHD.[5] However, regions that have a high unmet need, like the RGV, also face complex barriers to effective rapid care delivery.[6,7]

Quantitative Methods

- People were eligible if aged ≥18 years, if they had a CD4 count ≤200 cells/mm³ and/or CD4% ≤14% or diagnosed opportunistic infections or other AIDS-defining conditions, and if they initiated B/F/TAF treatment ≤7 days since diagnosis/referral to the clinic.
- The primary endpoint was 24 weeks, with a secondary endpoint of 48 weeks from B/F/TAF initiation. Follow-up was conducted until 24 weeks after the last person was enrolled in the study.
- Time to viral suppression was estimated using Kaplan-Meier analysis, and change from baseline analyses were conducted using the Wilcoxon Signed-Rank Test.

Qualitative Methods

- Internal policy and procedure documents were analyzed to develop a theory of change (**Figure 1**).
- Semi-structured interviews were performed with:
 1. ART-naïve, newly diagnosed adults with AHD
 2. Adults who were initiated onto B/F/TAF within 7 days from AHD diagnosis and were then receiving ongoing care
 3. Staff providing clinical and social services as part of the rapid start program
- Sampling was purposive and designed to maximize diversity. Sample size was determined by theoretical saturation.
- Interviews were conducted by peer researchers in English and Spanish. Results were transcribed and analyzed using deductive thematic analysis.[8]

Quantitative Results

Participant Demographics

- A total of 22 individuals were enrolled in the quantitative component.
 - One participant left the study before the first follow-up visit at four weeks from baseline, one participant was lost to follow-up after their Week 12 visit, and one participant switched off B/F/TAF after their Week 24 visit due to personal concerns regarding weight gain. Available data for all participants were included in the analysis.
- Baseline characteristics are reported in **Table 1**.
- No patients discontinued treatment due to virologic failure or resistance.

Time to Viral Suppression

- The median time to achieve an HIV-1 RNA viral load <200 copies/mL from baseline was 5.29 (95% CI: 4.57, 11.0) weeks (**Figure 2**).
- The median time to achieve an HIV-1 RNA viral load <50 copies/mL from baseline was 16.86 (95% CI: 13.29, 25.14) weeks.

Change from Baseline Analyses

- The median differences in HIV-1 RNA copies/mL, CD4 cell count (cells/mm³), CD4%, and CD4:CD8 ratio from baseline to 24 weeks and from baseline to 48 weeks are reported in **Table 2**.

Qualitative Results

Respondent Demographics

- 32 semi-structed interviews were conducted; 27 transcripts were included in the analysis, with 9 per subgroup. Of the 5 transcripts excluded, 3 were of poor quality, 1 individual was re-interviewed, and 1 audio recording failed.
- Among people with AHD, 7/18 interviews were conducted in Spanish.
- Interviews with staff included case managers (n=3), nurses (n=2), physicians (n=2), a rapid start pathway navigator, and a risk reduction specialist.
- Key themes are summarized below. Supporting quotes are provided in **Figure 3**.

Reasons for Late Diagnosis

- Reasons for late diagnosis included:
 - Limited knowledge about HIV prior to diagnosis (n=9), including a low perceived risk.
 - Psychological factors (n=9), such as the anxiety around the implications of a positive test result.
 - Medical oversight (n=8), often linked to primary care provider's lack of HIV knowledge.
 - Indistinguishable symptoms (n=7), often confused with other diseases such as COVID-19.
 - Poor access to HIV testing (n=3), including geographical barriers to testing in rural areas of Mexico and financial barriers.

Strengths and Challenges of Longer-Term Care

- Strengths of the approach for maximizing longer-term care engagement:
 - Tailored education and effective treatment that led to improvements in participants' physical and mental well-being.
 - Flexibility, adherence assessments, and frequent reminders for clinic appointments.
 - One individual expressed concern with the frequency of contact, viewing it as a threat to safeguarding their HIV status.
 - Free medication samples and support with insurance enrollment and optimization.
 - Transportation assistance.
- A key challenge that was not addressed by the program was transiency, with individuals lost to follow-up because they relocated, were deported or incarcerated.

FIGURE 1 Overview of patient intake and follow-up

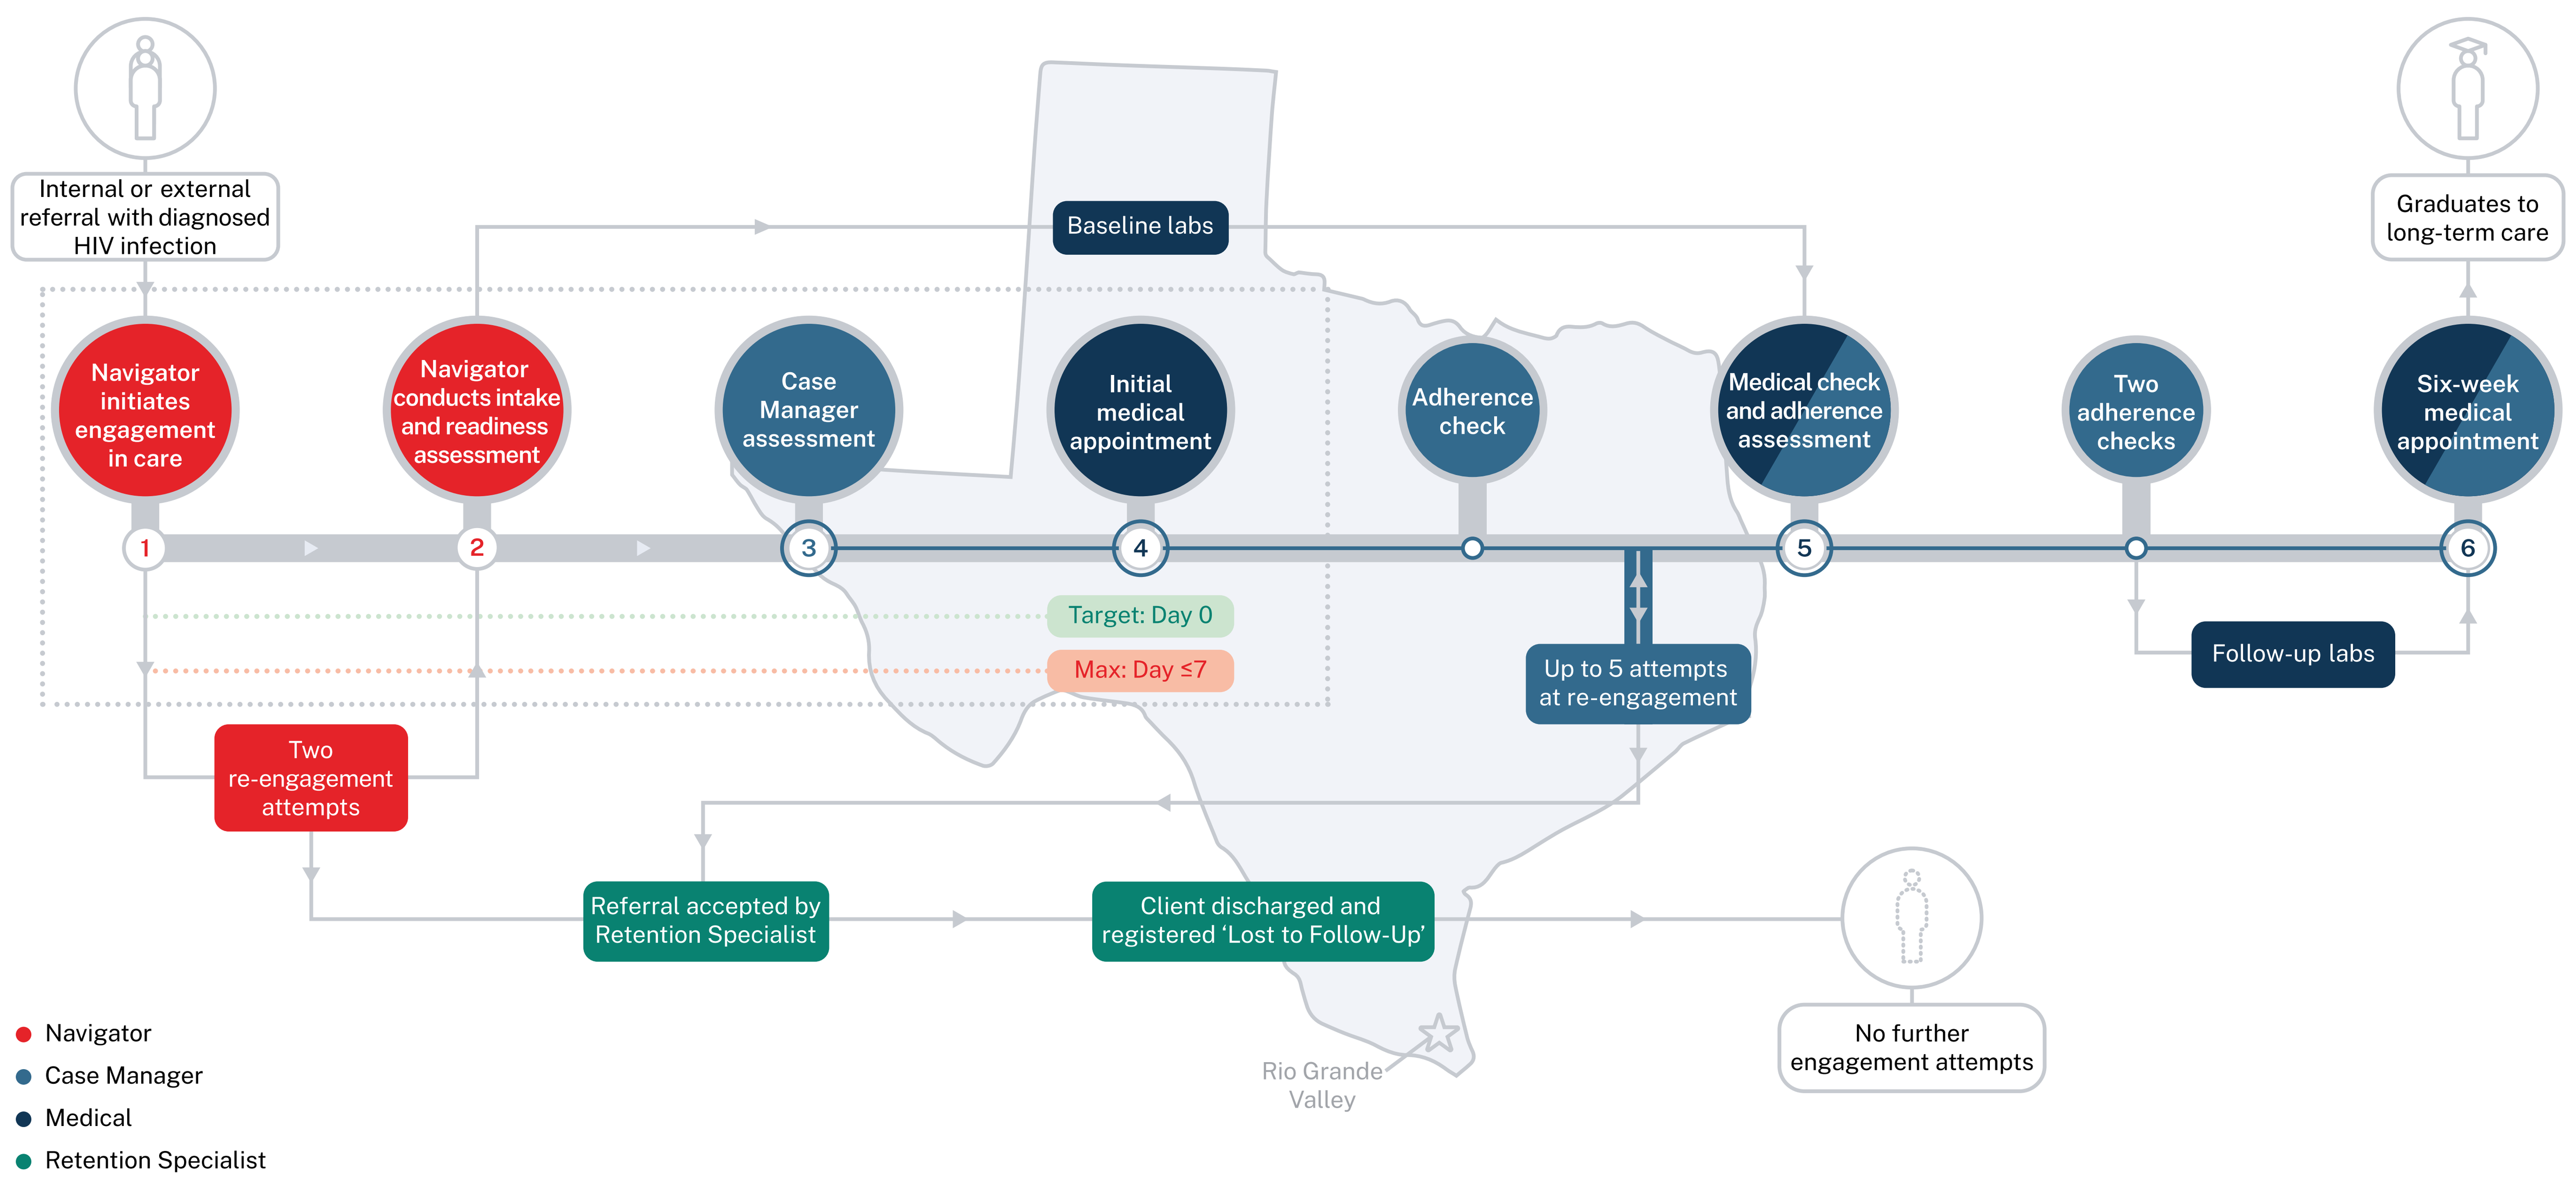
Internal or external referral with diagnosed HIV infection
Navigator initiates engagement in care
Navigator conducts intake and readiness assessment
Case Manager assessment
Initial medical appointment
Adherence check
Medical check and adherence assessment
Two adherence checks
Six-week medical appointment
Graduates to long-term care
Baseline labs
1
2
3
4
5
6
Target: Day 0
Max: Day ≤7
Up to 5 attempts at re-engagement
Follow-up labs
Two re-engagement attempts
Referral accepted by Retention Specialist
Client discharged and registered 'Lost to Follow-Up'
No further engagement attempts
Rio Grande Valley
Navigator
Case Manager
Medical
Retention Specialist

TABLE 1 Baseline characteristics

Baseline characteristics	Overall (N=22)
Age (years)	
Mean (SD)	39.6 (11.7)
Median [min, max]	37.9 [21.1, 60.4]
Sex, n (%)	
Male	20 (90.9%)
Female	2 (9.1%)
Sexual orientation, n (%)	
Homosexual	13 (59.1%)
Bisexual	2 (9.1%)
Heterosexual	7 (31.8%)
Race, n (%)	
American Indian or Alaska Native	0 (0.0%)
Asian	0 (0.0%)
Black or African American	0 (0.0%)
Native Hawaiian or Other Pacific Islander	0 (0.0%)
White	22 (100.0%)
Ethnicity, n (%)	
Hispanic/Latino	22 (100.0%)
Not Hispanic/Latino	0 (0.0%)
Insurance status, n (%)	
Medicare	1 (4.5%)
Medicaid	0 (0.0%)
Private	10 (45.5%)
Uninsured	11 (50.0%)
Other	0 (0.0%)
ADAP enrollment status, n (%)	
Yes	11 (50.0%)
No	11 (50.0%)
Housing status, n (%)	
Stable	22 (100.0%)
Unstable	0 (0.0%)
Annual Household Income ($)	
Mean (SD)	20,600 (20,400)
Median [min, max]	15,400 [0, 61,200]
Employment status, n (%)	
Employed	13 (59.1%)
Unemployed	9 (40.9%)

FIGURE 2 Time from B/F/TAF initiation to HIV-1 RNA <200 copies/mL

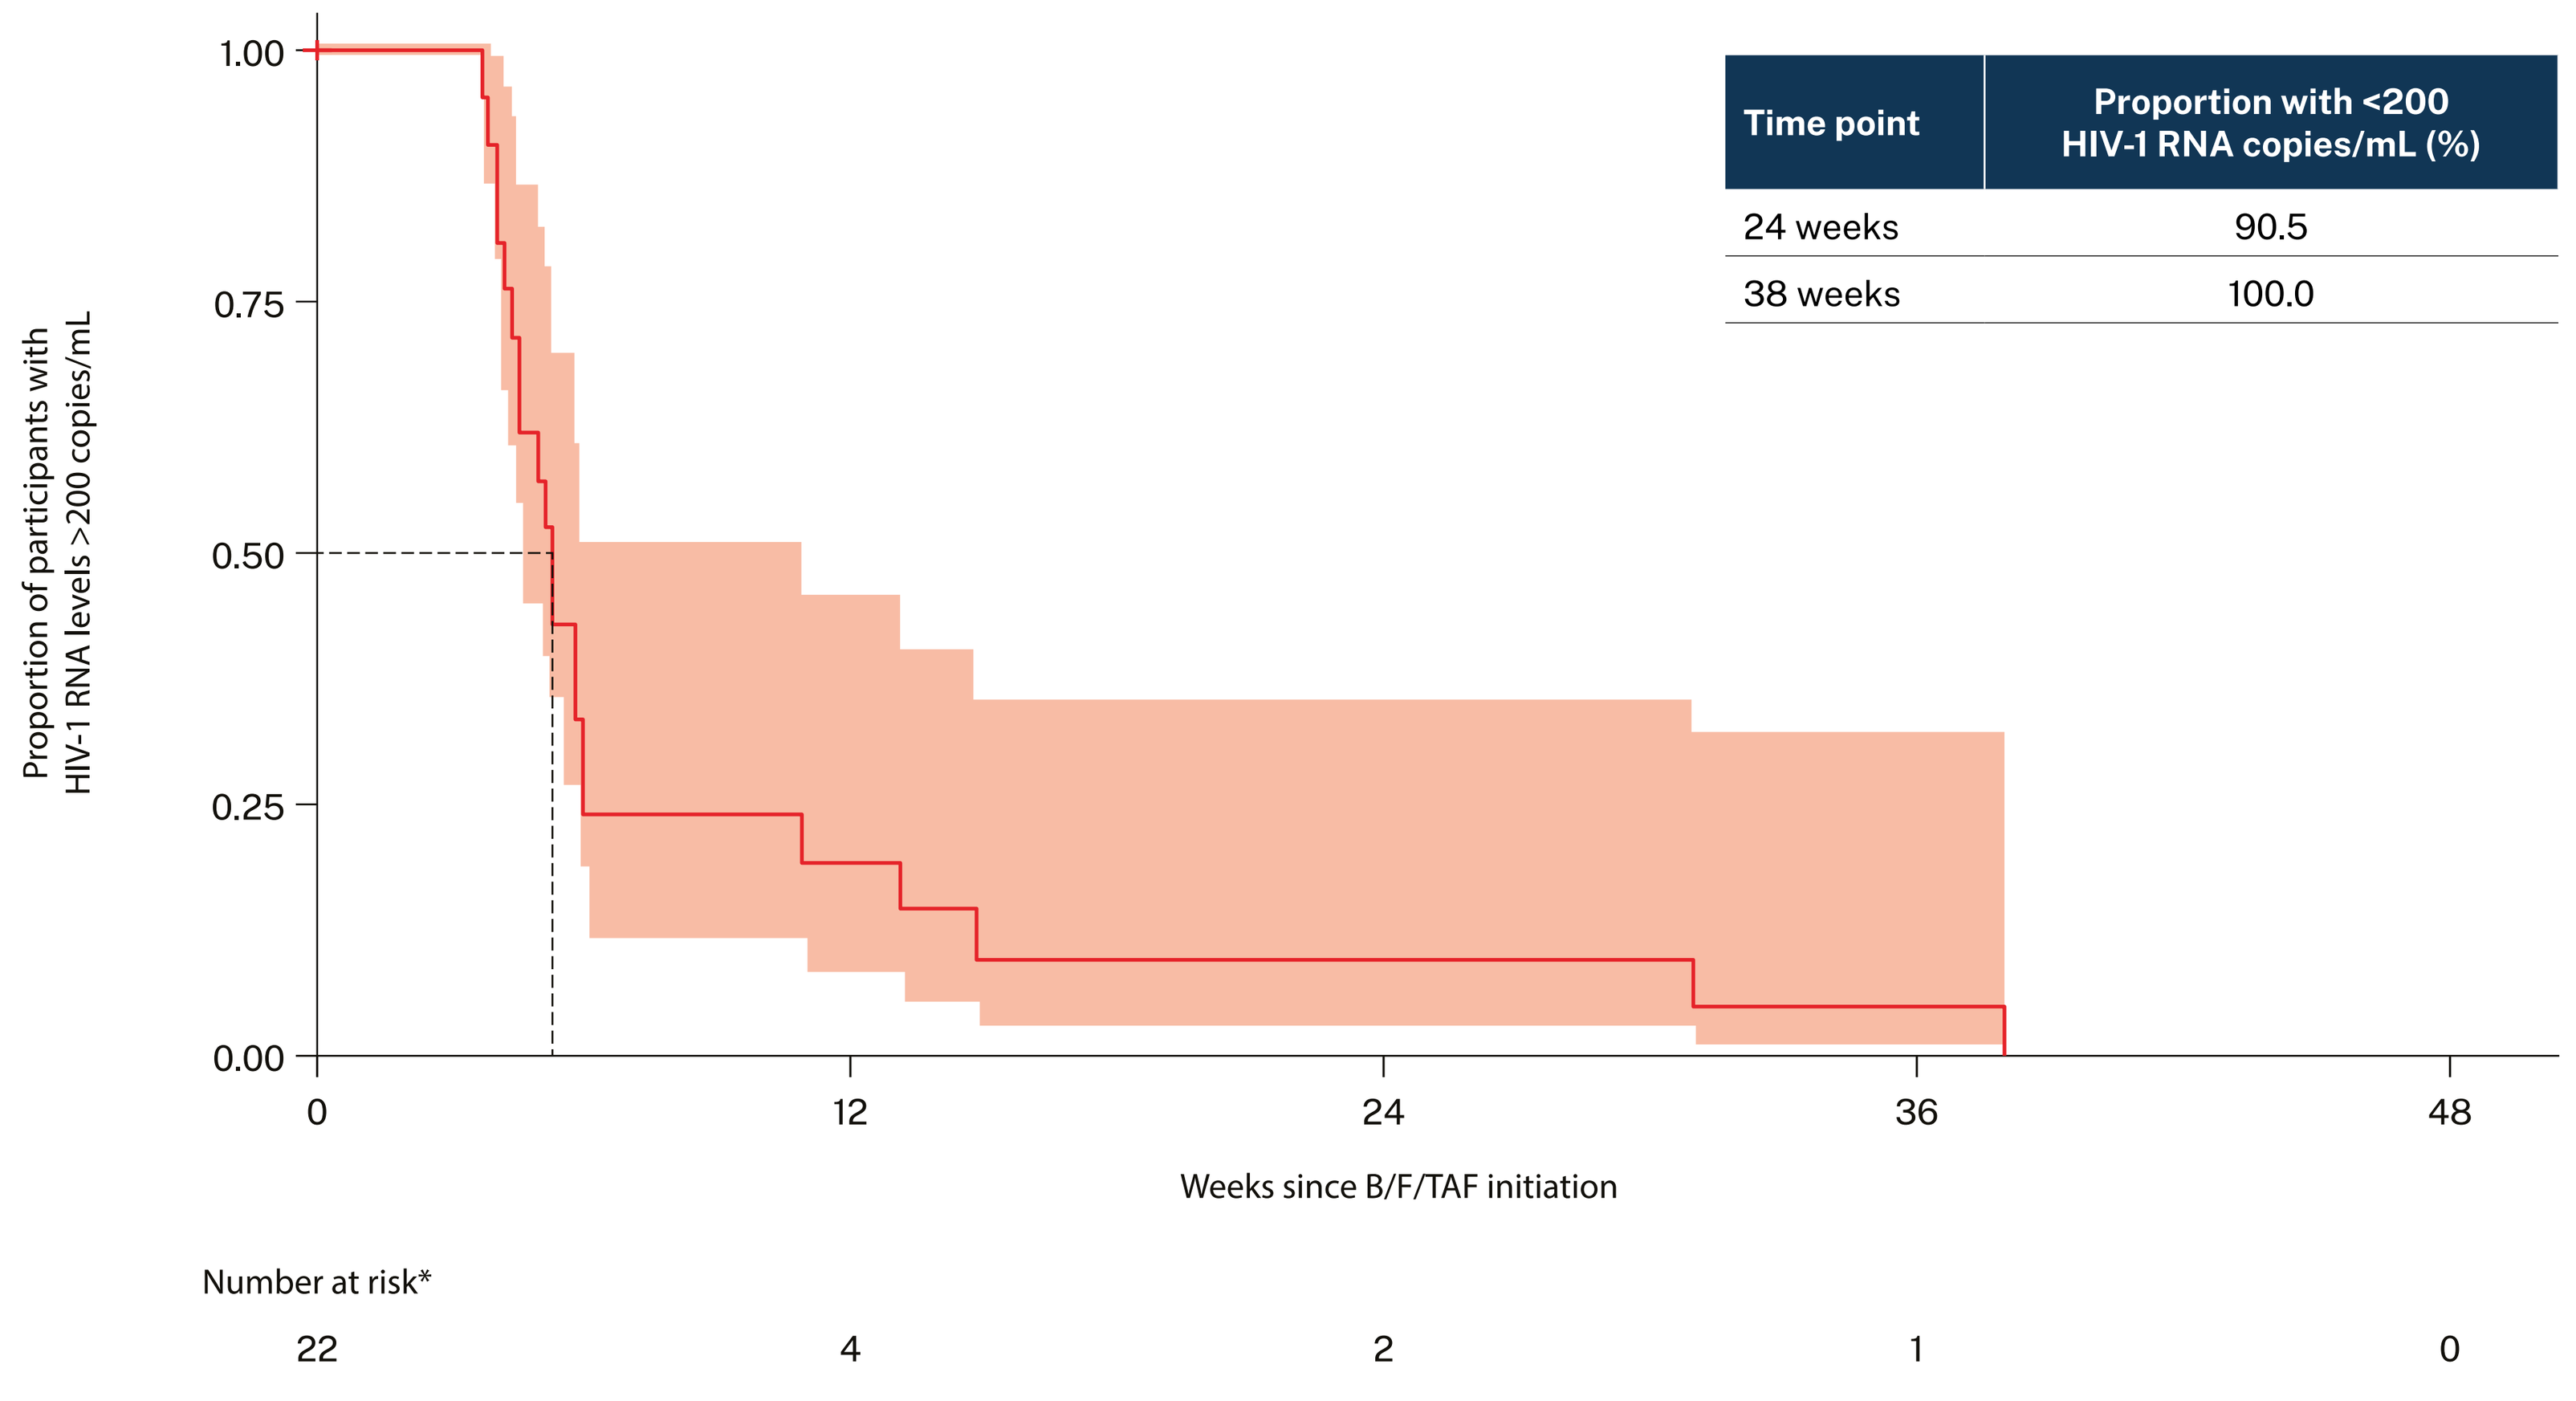
Proportion of participants with HIV-1 RNA levels >200 copies/mL
1.00
0.75
0.50
0.25
0.00
0
12
24
36
48
Weeks since B/F/TAF initiation
Number at risk*
22
4
2
1
0

Time point	Proportion with <200 HIV-1 RNA copies/mL (%)
24 weeks	90.5
38 weeks	100.0

*Number at risk refers to the number of participants at each timepoint who are still being observed and have not yet experienced the event of interest (achieving HIV-1 RNA level <200 copies/mL).

TABLE 2 Change in viral load and immune function recovery outcomes from B/F/TAF initiation

Outcome	Median difference from baseline to Week 24† visit	95% CI	Median difference from baseline to Week 48‡ visit	95% CI
HIV-1 RNA copies/mL ($\log_{10}$)	-3.7***	-4.1, -3.2	-3.8***	-4.5, -3.2
CD4 count (cells/mm³)	126**	68, 246	207***	138, 266
CD4% (%)	5.9***	4.4, 7.8	9.2***	6.7, 12.0
CD4:CD8 ratio	0.1***	0.1, 0.3	0.3**	0.2, 0.5

†Mean (SD) weeks from baseline to Week 24 visit: 24.1 (3.0); ‡Mean (SD) weeks from baseline to Week 48 visit: 51.5 (6.2); **p<0.01; ***p<0.001.

FIGURE 3 Sample qualitative data

Reasons for late diagnosis

Physical barriers

"…**there are [HIV services], but it's like I tell you**, maybe because we are coming from a different area and it's more focused in the middle of the city and because of the jobs that we have [outside of the city], they don't give you permission to go."
41 y.o. male, diagnosed and receiving care

Financial barriers

"…**most of our patients do not have health insurance**. So they're coming from a low income, you know low resource area where most of their employment doesn't offer health insurance […] so the only time that they go to the doctors is when they're really, really sick and they go to the ER [emergency room]…"
42 y.o. male, rapid start staff

Stigma

"**Over there in my country I never had the intention of testing** until I got here, with the fear that my family would say this or that or my friends you know […] I've always heard that the US is a country of many opportunities and that it lends help to those from other countries, so I chose [to test] and I looked [for clinics] and until this day I am here, thank God."
41 y.o. male, diagnosed and receiving care

Myths and misconceptions

"…**I thought it was a gay thing**. I didn't think it was like, like I would never get it, I would never get it and I was dead wrong. I mean I got it from a homeless girl on the streets and she told me she had leukemia, cancer, she was dying and I felt sorry for her […] in fact, she had full blown AIDS, she died like a month later…"
37 y.o. male, newly diagnosed

"…he would call me on the phone and he would ask me, have you tested yourself [for HIV]? And I would say yes, and he would ask what the result was. **But I hadn't tested myself because at that time I wasn't feeling ill**…"
51 y.o. male, diagnosed and receiving care

Strengths and challenges of longer-term care

Supportive scheduling

"Yeah, um I guess **whenever I have an appointment I might not always remember so they'd actually call me too, in advance the day before** to, to see if I'm gonna make it to the appointment. And luckily when they do, I can tell them whether I'm not gonna make it or whether I need transportation in advance."
36 y.o. male, diagnosed and receiving care

Sustained access

"Well, the only barrier, maybe, would have been transportation maybe […] but they were able to, they were able to assist me with it […] **they sent me an Uber all the way to [city] for my eye exam** and then also, they-they picked me up a couple of times from my apartment in [city]…"
36 y.o. male, diagnosed and receiving care

Effective treatment

"So, at first, I was like, my first instinct was like, 'oh I'm gonna die' […] I was like 'I'm not going to make it much [more]' and then, **well that's when I started coming in and they gave me the medication […] that's when I started seeing a difference**, like, 'okay never mind I'm going to live a little bit more' (laughs), 'a lot more', so yeah, [the medication's] been great."
30 y.o. male, newly diagnosed

Transiency

"…we have a lot of people that come from across the border from Mexico, you know, and they may be here, you know, illegally and, you know, **they may get deported or, you know, end up in the prison system**. […] we have those migrant patients that, you know, cross back and forth […] we've tried to coordinate care with CAPASITS[a] in Mexico, but that hasn't really worked…"
42 y.o. male, rapid start staff

[a]Centro Ambulatorio para la Prevención y Atención en SIDA e Infecciones de Transmisión Sexual. Translates to Outpatient Center for the Prevention and Care of AIDS and Sexually Transmitted Infections.

Conclusions

- **Rapid ART initiation is effective in a resource-constrained Latine community of people with AHD when using B/F/TAF before baseline lab results, combined with a holistic healthcare approach that emphasizes HIV education, psychosocial and logistical support, and reduces economic barriers.**

Abbreviations: **ADAP:** AIDS Drug Assistance Program; **AHD:** advanced HIV disease; **ART:** antiretroviral therapy; **B/F/TAF:** bictegravir/emtricitabine/tenofovir alafenamide; **CI:** confidence interval; **RGV:** Rio Grande Valley; **SD:** standard deviation.

References: [1]Creswell, JW; Plano Clark, VL. Designing and Conducting Mixed-Methods Research: SAGE Publications Inc.; 2018; [2]Unidos Contra la Diabetes. Rio Grande Valley 2024 Demographics. 2024. Available from: https://www.rgvhealthconnect.org/index.php?module=DemographicData&controller=index&action=index. Last accessed: 4/15/2025; [3]Texas Department of State Health Services, TDoSH. Texas HIV Surveillance Report. 2022; [4]De La Garza, R; Rodrigo, H; Fernandez, F et al. The Increase of HIV-1 Infection, Neurocognitive Impairment, and Type 2 Diabetes in The Rio Grande Valley. Curr HIV Res. 2019;17(6):377–387; [5]Panel on Antiretroviral Guidelines for Adults and Adolescents. Guidelines for the Use of Antiretroviral Agents in Adults and Adolescents with HIV. Department of Health and Human Services. Available from: https://clinicalinfo.hiv.gov/en/guidelines/adult-and-adolescent-arv. Last accessed: 10/2024; [6]Guilamo-Ramos, V; Thimm-Kaiser, M; Benzekri, A et al. The Invisible US Hispanic/Latino HIV Crisis: Addressing Gaps in the National Response. American Journal of Public Health. 2020;110(1):22–68; [7]Servin, AE; Muñoz, FA; Zúñiga, ML. Healthcare provider perspectives on barriers to HIV-care access and utilisation among Latinos living with HIV in the US-Mexico border. Culture, Health & Sexuality. 2014;16(5):587–599; [8]Braun, V; Clarke, V. Using thematic analysis in psychology. Qualitative Research in Psychology. 2006;3(2):77–101. **Acknowledgements:** The authors acknowledge Dr. Dora Martinez (MD, FAAFP, AAHIVS) for support with study conceptualization. We also extend our gratitude to everyone who contributed to the success of this study, including all the participants and their families, Valley AIDS Council clinic staff, and the peer researchers for their invaluable support. This study was funded by Gilead Sciences. Editorial services provided by Costello Medical and funded by Gilead Sciences. **Disclosures:** **BN** and **SM** are employees of Gilead and hold stock interest in the company.

The American Conference for the Treatment of HIV (ACTHIV) | Chicago, IL, USA | 1–3 May 2025