17

Switch to DOR/ISL (100/0.25 mg) QD from BIC/FTC/TAF: 96-Week Update from a Blinded Phase 3 Study

Amy E. Colson, MD, MPH[1]; Anthony M. Mills, MD[2]; Moti Ramgopal, MD[3]; Christopher Bettacchi, MD[4]; Olayemi Osiyemi, MD[5]; Federico Hinestrosa, MD[6]; Gordon Crofoot, MD[7]; Harold P. Katner, MD[8]; Hiroyuki Gatanaga, MD[9]; Margaret Johnson, MD[10]; Chenguang Zhang, PhD[11]; Tracy L. Diamond, PhD[11]; Erika Barninger, BS[11]; Michelle C. Fox, MD[11]; Mengchun Li, MD[11]; Chikelue Oragwu, MD*

[1]Community Resource Initiative, Boston, MA and Cambridge Health Alliance, Cambridge, MA, USA; [2]Men's Health Foundation, Los Angeles, CA, USA; [3]Midway Immunology and Research Center, Fort Pierce, FL, USA; [4]HIV Center, North Texas Infectious Diseases Consultants, Dallas, TX, USA; [5]Triple O Research Institute, West Palm Beach, FL, USA; [6]Orlando Immunology Center, Orlando, FL, USA; [7]The Crofoot Research Center, Inc., Houston, TX, USA; [8]Mercer University School of Medicine, Macon, GA, USA; [9]AIDS Clinical Center, NCGM, Tokyo, Japan; [10]Royal Free London NHS Foundation Trust, London, UK; [11]Merck & Co., Inc., Rahway, NJ, USA

*Presenting on behalf of the authors

Background

- Given the longer life expectancy and high prevalence of age-related comorbidities in people living with HIV-1, there is an unmet need for effective and tolerable two-drug regimens that minimize the risk of drug-disease and drug-drug interactions and limit lifetime exposure to antiretroviral medication[1]
- The combination of doravirine (DOR) and islatravir (ISL) shows promise as a simplified oral once-daily (QD) treatment option for HIV-1
 - DOR is a non-nucleoside reverse transcriptase inhibitor (NNRTI) with activity against common NNRTI-resistant variants and with a favorable safety profile[2]
 - ISL is an investigational nucleoside reverse transcriptase translocation inhibitor (NRTTI) with multiple mechanisms of action, potent in vitro antiviral activity (including against common NRTI-resistant variants), and a high barrier to the emergence of resistance[3,4]
 - DOR and ISL have complementary resistance profiles; variants encoding the most common DOR resistance-associated mutation observed in DOR/3TC/TDF Phase 3 studies are hypersusceptible to ISL[3]
- MK8591A-052 is a double-blind study evaluating the efficacy and safety of DOR/ISL in virologically suppressed people living with HIV-1
 - Switching to DOR/ISL 100/0.25 mg was noninferior to continuing bictegravir/emtricitabine/tenofovir alafenamide (BIC/FTC/TAF) based on the primary endpoint of participants with HIV-1 RNA ≥50 copies/mL at Week 48[5], observed in 1.5% of DOR/ISL versus 0.6% of BIC/FTC/TAF participants (treatment difference 0.9%; 95% confidence interval [CI]: -1.9, 2.9)
 - DOR/ISL and BIC/FTC/TAF also had similar adverse event (AE) profiles through Week 48[5]
- Here we present Week 96 efficacy and safety data from this ongoing study

Methods

- MK8591A-052 (NCT05630755) is a 2:1 randomized, active-controlled, double-blinded, multicenter, Phase 3 non-inferiority study (**Figure 1**)[5]
- Primary analysis at Week 48 demonstrated that switching to DOR/ISL had non-inferior efficacy and comparable safety to continuing BIC/FTC/TAF[5]
- Secondary efficacy endpoints assessed at Week 96 (no formal hypothesis testing) included: HIV-1 RNA ≥50 copies/mL (FDA snapshot), HIV-1 RNA <50 and <200 copies/mL, and CD4+ T-cell counts
- Post-baseline resistance analysis was performed for participants with confirmed HIV-1 RNA ≥200 copies/mL and those who discontinued with HIV-1 RNA ≥200 copies/mL

Figure 1. Double-blind DOR/ISL vs continuing BIC/FTC/TAF

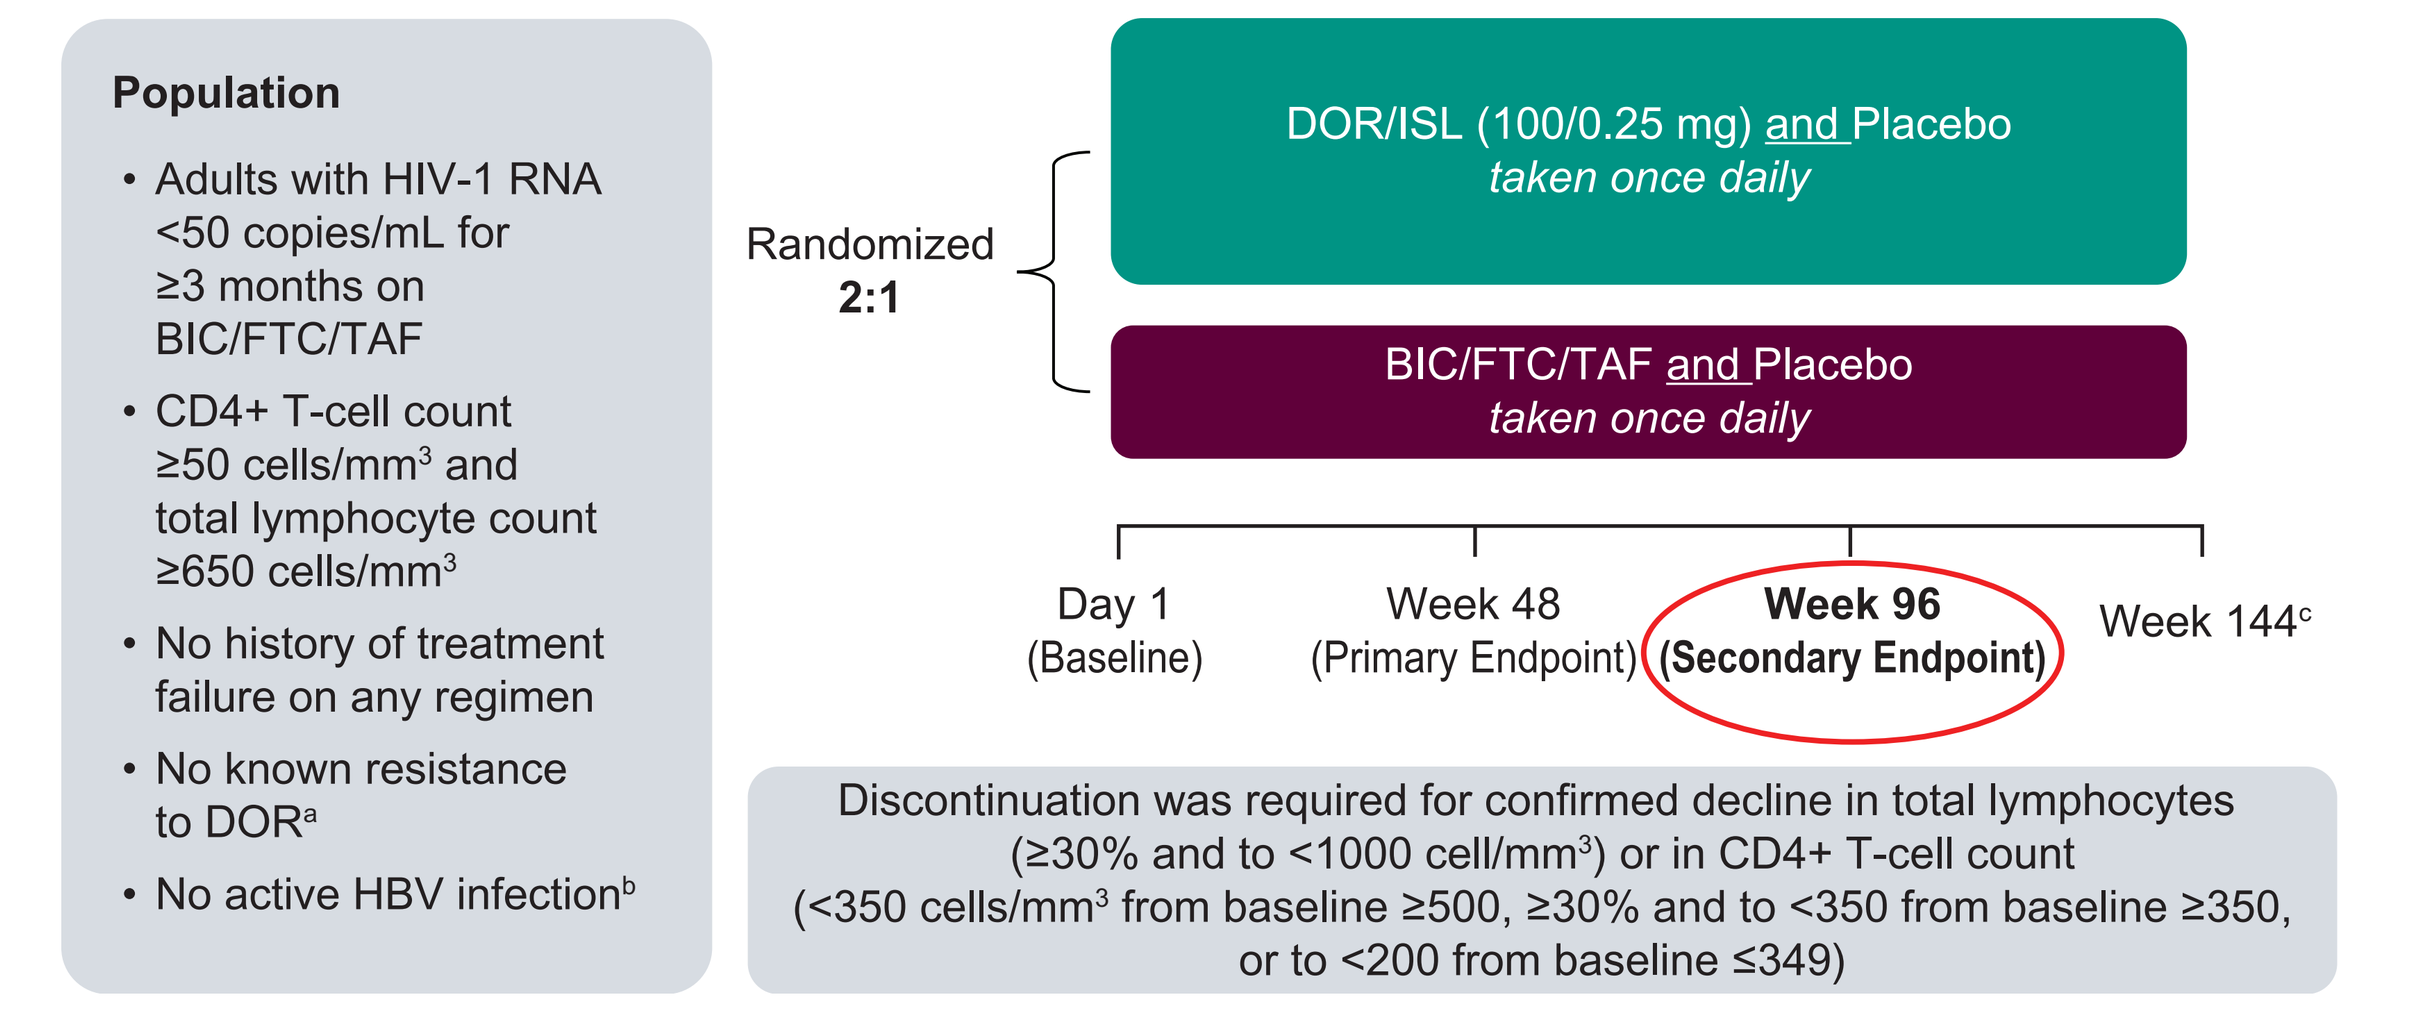
Population
Adults with HIV-1 RNA <50 copies/mL for ≥3 months on BIC/FTC/TAF
CD4+ T-cell count ≥50 cells/mm³ and total lymphocyte count ≥650 cells/mm³
No history of treatment failure on any regimen
No known resistance to DOR[a]
No active HBV infection[b]
Randomized 2:1
DOR/ISL (100/0.25 mg) and Placebo
taken once daily
BIC/FTC/TAF and Placebo
taken once daily
Day 1 (Baseline)
Week 48 (Primary Endpoint)
Week 96 (Secondary Endpoint)
Week 144[c]
Discontinuation was required for confirmed decline in total lymphocytes (≥30% and to <1000 cell/mm³) or in CD4+ T-cell count (<350 cells/mm³ from baseline ≥500, ≥30% and to <350 from baseline ≥350, or to <200 from baseline ≤349)

Specific study sponsor staff involved in data analysis and interpretation were unblinded to treatment assignment at Week 48. The participants, investigators, site staff, and sponsor field team will remain blinded through Week 144.
[a]V106A/M, V108I, Y188L, H221Y, P225H, F227C/L/V, M230I/L, L234I, P236L, and/or Y318F. [b]Defined as negative surface antigen and DNA tests. [c]After unblinding at Week 144, participants from both groups may enter an open-label extension phase receiving DOR/ISL 100/0.25 mg for up to 96 weeks (or until local approval and commercial availability).

Results

- As previously reported,[5] 342 participants switched to DOR/ISL and 171 continued BIC/FTC/TAF
- The study enrolled a diverse population (**Table 1**)

Table 1. Baseline characteristics were comparable between groups

Characteristic	Total Population N = 513	DOR/ISL N = 342	BIC/FTC/TAF N = 171
Age (years), median (IQR)	47.0 (37.0 – 58.0)	48.0 (37.0 – 60.0)	47.0 (37.0 – 58.0)
Female sex at birth, n (%)	110 (21.4)	74 (21.6)	36 (21.1)
Race, n (%): Black or African American	158 (30.8)	111 (32.5)	47 (27.5)
White	312 (60.8)	206 (60.2)	106 (62.0)
Asian	30 (5.8)	20 (5.8)	10 (5.8)
Ethnicity, n (%): Hispanic or Latino	117 (22.8)	73 (21.3)	44 (25.7)
Years since HIV-1 diagnosis, median (IQR)	11.0 (4.0 – 19.0)	11.3 (4.0 – 19.0)	10.6 (5.0 – 19.0)
Years of prior BIC/FTC/TAF, median (IQR)	3.4 (2.0 – 5.0)	3.3 (2.0 – 5.0)	3.6 (2.0 – 5.0)
CD4+ T-cell count, median (IQR) cells/mm³	691 (499 – 882)	681 (497 – 874)	705 (512 – 920)
CD4+ T-cell count ≥350 cells/mm³, n (%)	466 (90.8)	315 (92.1)	151 (88.3)

IQR, interquartile range; n, number of participants meeting the corresponding row criterion; N, total number of participants.

- At Week 96, confirmed HIV-1 RNA ≥50 copies/mL occurred in 5 (1.5%) participants in the DOR/ISL compared with 2 (1.2%) in the BIC/FTC/TAF group (**Figure 2**)
- HIV-1 RNA <50 copies/mL (per FDA snapshot approach) was maintained in 88.9% in the DOR/ISL and 90.1% in the BIC/FTC/TAF group and <200 copies/mL in 88.9% and 91.2%, respectively (**Figure 2**)
- Through Week 96, 2 (0.6%) participants in the DOR/ISL and 0 in the BIC/FTC/TAF group discontinued due to lack of efficacy; 3 (0.9%) vs 0, respectively, discontinued for other reasons with HIV-1 RNA ≥50 copies/mL at discontinuation

Figure 2. Efficacy at Week 96 (FDA snapshot approach) was similar between DOR/ISL and BIC/FTC/TAF

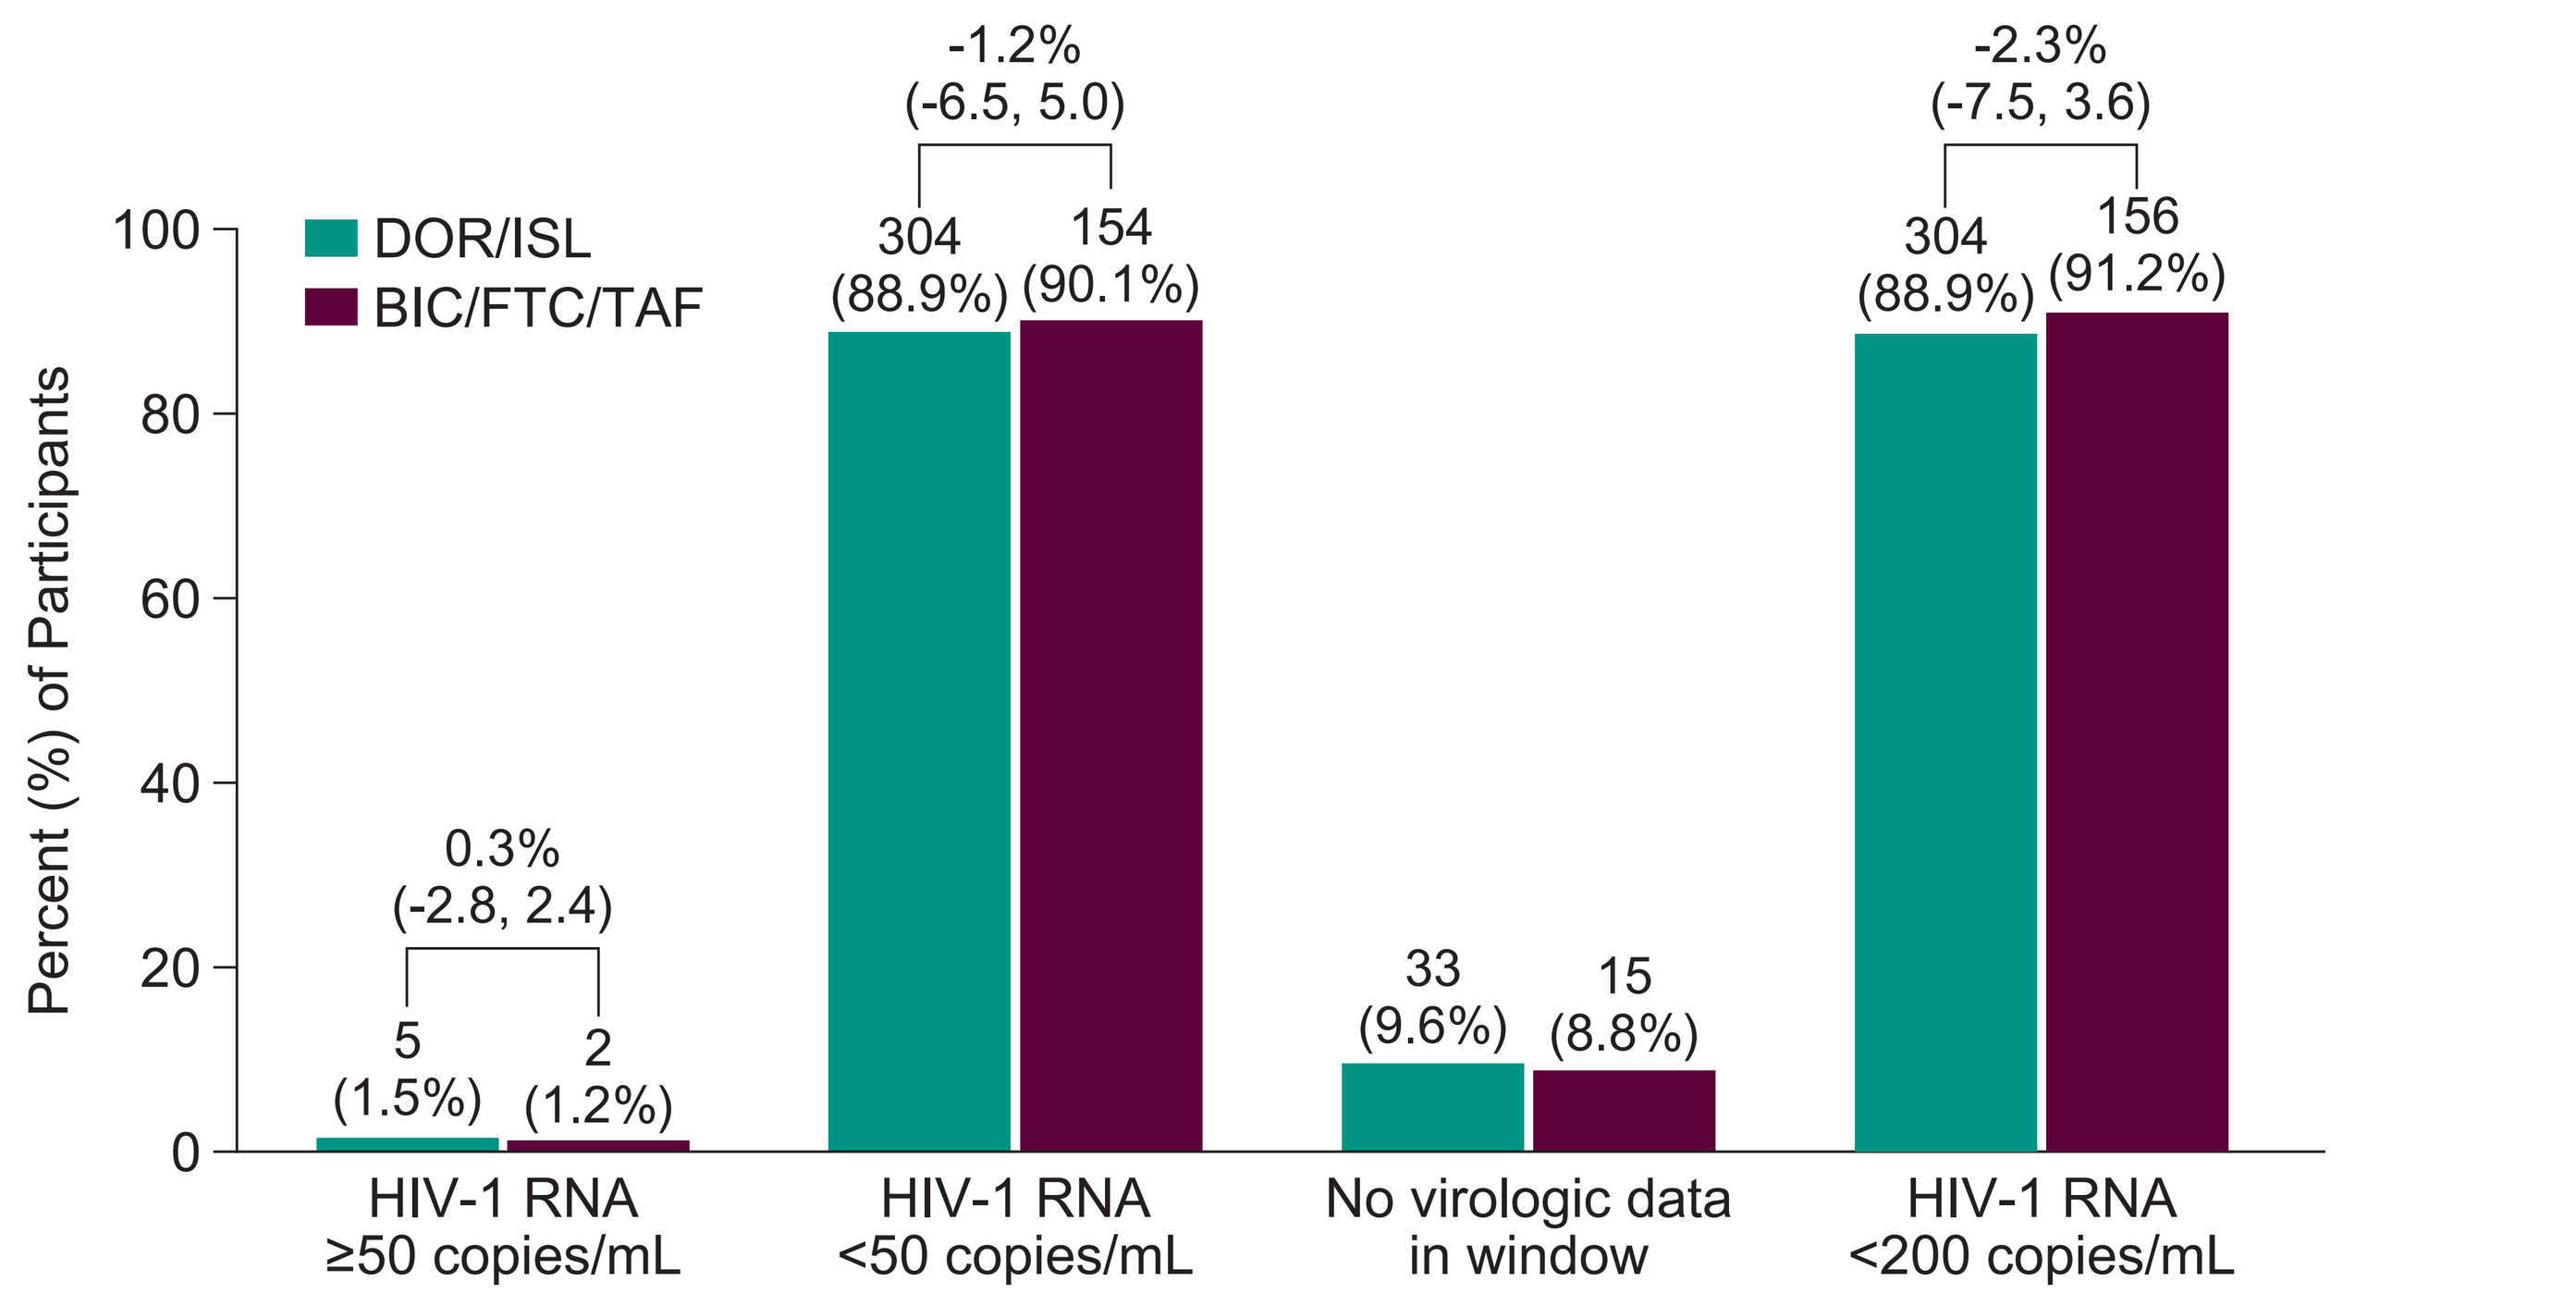
DOR/ISL
BIC/FTC/TAF
Percent (%) of Participants
100
80
60
40
20
0
0.3%
(-2.8, 2.4)
5
(1.5%)
2
(1.2%)
-1.2%
(-6.5, 5.0)
304
(88.9%)
154
(90.1%)
33
(9.6%)
15
(8.8%)
-2.3%
(-7.5, 3.6)
304
(88.9%)
156
(91.2%)
HIV-1 RNA ≥50 copies/mL
HIV-1 RNA <50 copies/mL
No virologic data in window
HIV-1 RNA <200 copies/mL

- Through 96 weeks, treatment-emergent resistance was not detected in the BIC/FTC/TAF group versus in 1 out of 342 participants (0.3%) in the DOR/ISL group (in a post hoc analysis)
 - This participant had HIV-1 RNA >200 copies/mL at a single visit (Week 24) and withdrew consent at Week 27.5 with HIV-1 RNA <200 copies/mL (thus not meeting pre-specified criteria for resistance testing)
 - Treatment-emergent substitutions (H221Y, M230L, and L234I) were detected at Week 24 that conferred phenotypic resistance to DOR
 - M184V was detected at baseline (proviral DNA) and Week 24; the virus remained sensitive to ISL at Week 24 with 7.4-fold decreased susceptibility versus reference virus
- Mean percent changes (data as observed) in CD4+ T-cell counts and total lymphocyte counts (TLC) were comparable between groups (**Figure 3**)
- At Week 96, mean change from baseline in CD4+ T-cell counts was 21.9 (95% CI: 1.4, 42.5) cells/mm³ with DOR/ISL and 26.7 (-2.6, 55.9) cells/mm³ with BIC/FTC/TAF (difference: 1.6 cells/mm³; 95% CI: -31.5, 34.7)
- Mean change from baseline in TLC was -0.02 (95% CI: -0.07, 0.03) x10⁹ cells/L with DOR/ISL and 0.01 (95% CI: -0.05, 0.08) x10⁹ cells/L with BIC/FTC/TAF (difference: -0.03 x10⁹ cells/L; 95% CI: -0.10, 0.05)
- In both groups, 0.6% (DOR/ISL n=2, BIC/FTC/TAF n=1) discontinued due to protocol-specified decreases in CD4+ T-cell count and/or TLC

Figure 3. Mean percent changes in total lymphocyte and CD4+ T-cell counts

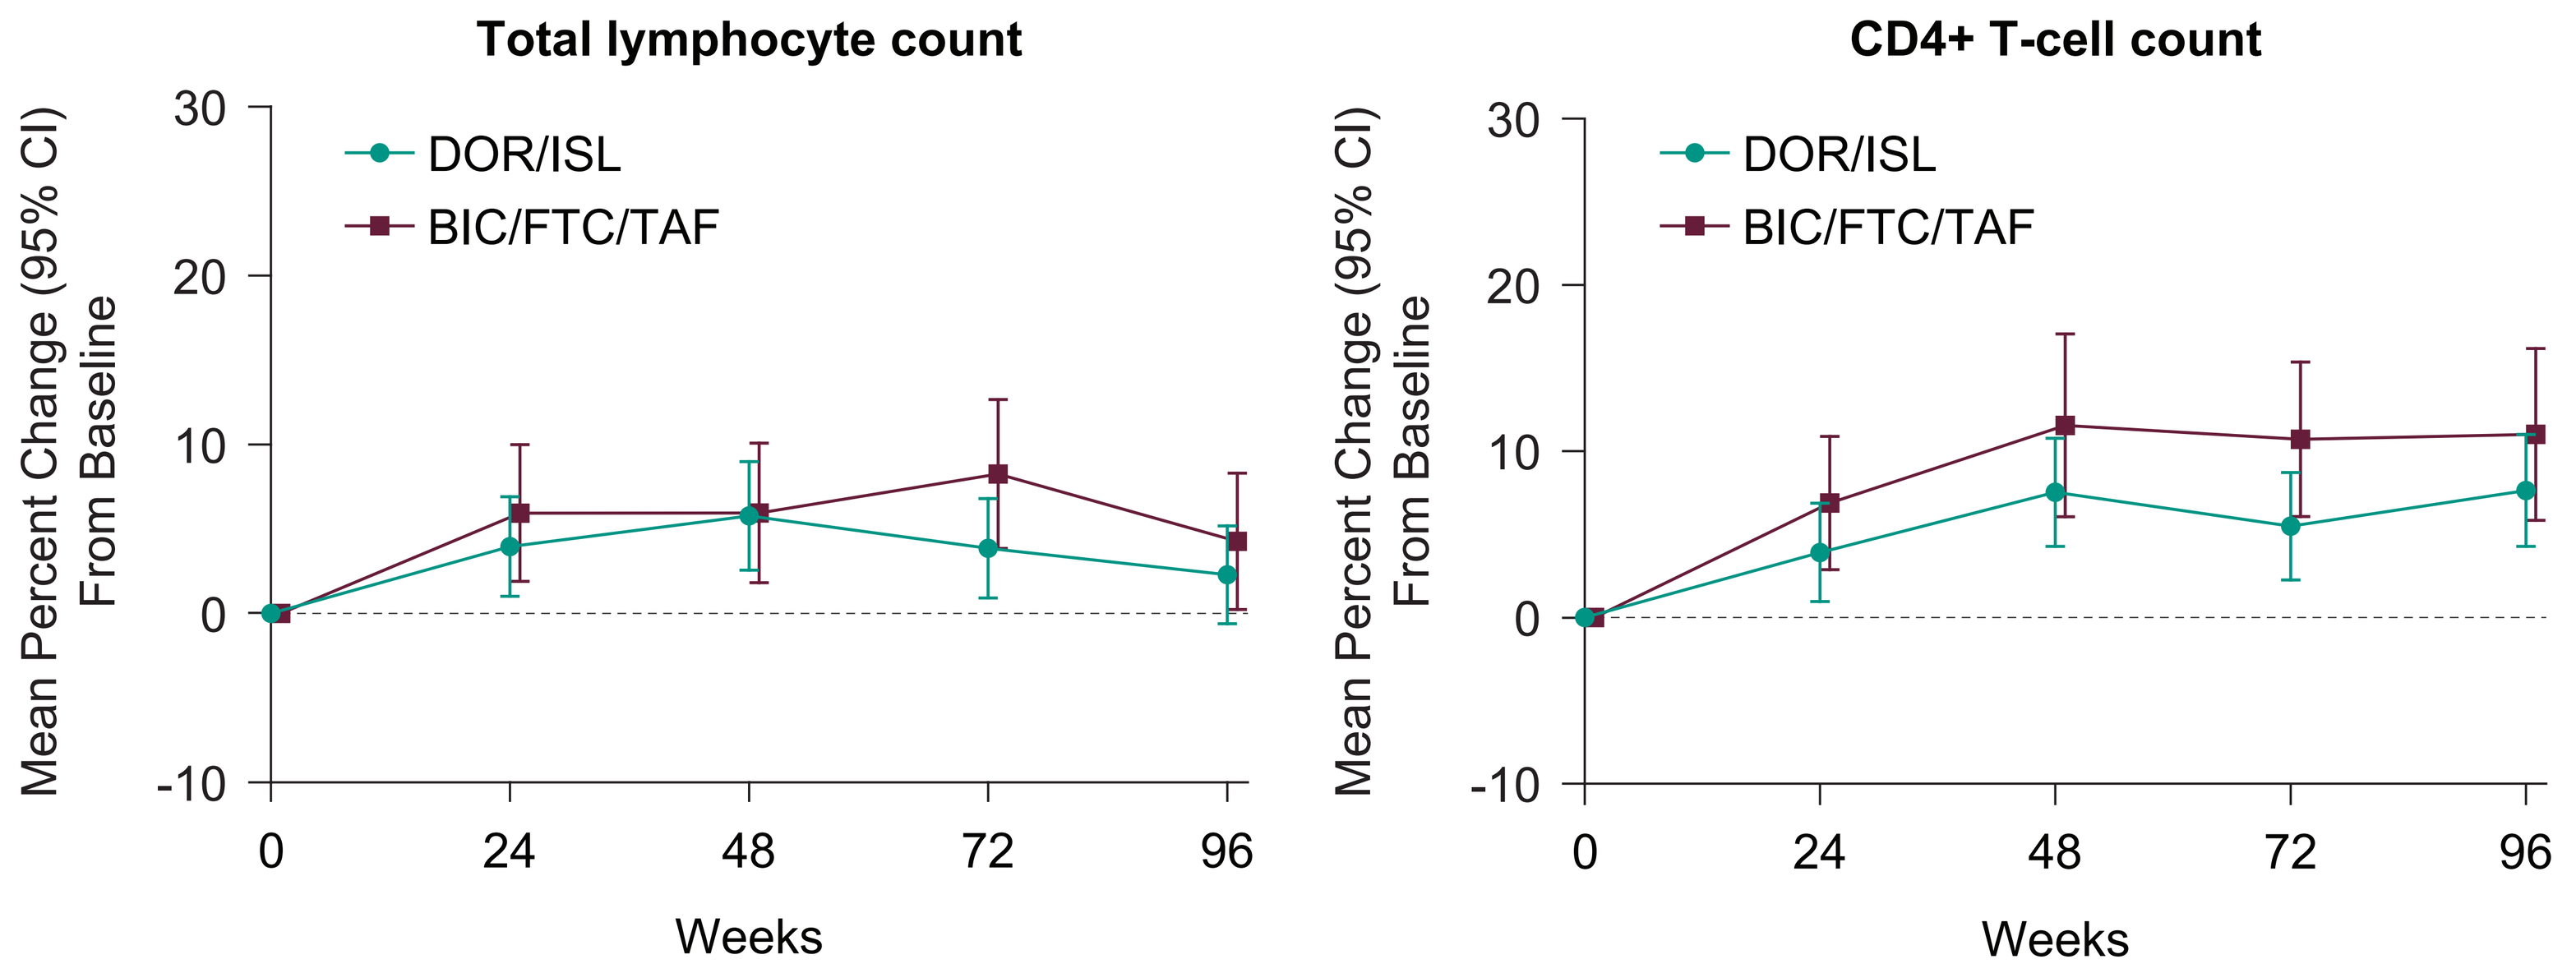
Total lymphocyte count
CD4+ T-cell count
DOR/ISL
BIC/FTC/TAF
Mean Percent Change (95% CI) From Baseline
30
20
10
0
-10
0
24
48
72
96
Weeks

- Proportions of participants with AEs through Week 96 were similar between groups (**Table 2**)
- The most common (>1%) drug-related AEs as assessed by the investigator were diarrhea (1.5%), flatulence, decreased lymphocyte count, and pruritis (1.2% each) with DOR/ISL and sleep disorder, osteopenia, decreased weight, and decreased appetite (1.2% each) with BIC/FTC/TAF

Table 2. The adverse event profiles of DOR/ISL and BIC/FTC/TAF were comparable

	DOR/ISL N = 342	BIC/FTC/TAF N = 171	Treatment Difference
	n (%)	n (%)	% (95% CI)[a]
With ≥1 AE	290 (84.8)	144 (84.2)	0.6 (-5.8, 7.7)
Drug-related[b] AEs	38 (11.1)	18 (10.5)	0.6 (-5.6, 6.0)
Grade 3 - 4 AEs	39 (11.4)	18 (10.5)	0.9 (-5.4, 6.3)
Serious AEs	28 (8.2)	15 (8.8)	-0.6 (-6.3, 4.2)
Drug-related[b] serious AEs	1 (0.3)[c]	0 (0)	0.3 (-1.9, 1.6)
Deaths	0 (0)	0 (0)	0.0 (-2.2, 1.1)
Discontinued due to an AE	11 (3.2)	5 (2.9)	0.3 (-3.7, 3.3)
Discontinued due to a drug-related[b] AE	5 (1.5)	4 (2.3)	-0.9 (-4.5, 1.5)
Discontinued due to a drug-related[b] serious AE	1 (0.3)[c]	0 (0)	0.3 (-1.9, 1.6)

[a]Based on the Miettinen & Nurminen method. [b]Considered by the investigator to be related to the study drug. [c]This participant discontinued treatment due to a drug-related serious AE of 'immune thrombocytopenia' at Week 4, as previously reported.[5]

- There were no cases of clinical HBV reactivation; transient low-level HBV viremia (DNA ≤36 IU/mL) was detected in 3 DOR/ISL participants HBcAb-positive at screening (**Table 3**), but none had symptoms, surface antigenemia, or transaminase elevation (2 viremias resolved on DOR/ISL without treatment, 1 participant discontinued DOR/ISL)
- New HBV infections (i.e., HBcAb-negative at screening followed by confirmed HBV DNA >10 IU/mL) occurred in 4 DOR/ISL participants (**Table 3**), all of whom remained asymptomatic
 - Two participants were HBV surface antigen positive with DNA >200 000 IU/mL and discontinued DOR/ISL; subsequently, HBV viremia resolved after initiation of an antiretroviral regimen with anti-HBV activity
 - Two participants had low-level HBV viremia (DNA <120 IU/mL), negative HBV surface antigen, and normal transaminases; HBV viremia resolved on DOR/ISL in both

Table 3. HBV events through Week 96 by screening serology

HBV Serology	DOR/ISL N = 342	BIC/FTC/TAF N = 171
HBcAb positive, HBsAb positive at screening, n (%)	74 (21.6)	40 (23.4)
HBV DNA detected, m (%)[a]	2 (2.7)	0 (0.0)
HBcAb positive, HBsAb negative at screening, n (%)	12 (3.5)	6 (3.5)
HBV DNA detected, m (%)[a]	1 (8.3)	0 (0.0)
HBcAb negative, HBsAb negative at screening, n (%)	103 (30.1)	50 (29.2)
New HBV infection, m (%)[b]	4 (3.9)	0 (0.0)

Hepatitis B serology (HBcAb, HBsAb, and HBV DNA) was assessed at screening, Week 48, and Week 96; HBsAg and HBV DNA were assessed at baseline and every 4 weeks thereafter in participants HBcAb-positive at screening.

[a]Confirmed HBV DNA >10 IU/mL in participants with positive baseline HBcAb. [b]Confirmed HBV DNA >10 IU/mL with negative baseline HBcAb.

HBcAb, hep B core antibody; HBsAb, hep B surface antibody; m, number of participants with the respective baseline serology and the respective HBV event; n, number of participants with serology results at screening; N, total number of participants in the respective treatment group.

Table 4. Mean changes from baseline to Week 96 in weight, lipids, insulin resistance, and renal function were comparable between groups

Measure	DOR/ISL n	DOR/ISL Mean change[a]	BIC/FTC/TAF n	BIC/FTC/TAF Mean change[a]	Treatment Difference (95% CI)[b]
Weight, kg	304	0.34 (-0.38, 1.06)	156	0.43 (-0.37, 1.24)	-0.03 (-1.18, 1.11)
Fasting LDL-C, mg/dL	289	-1.76 (-4.99, 1.47)	146	0.36 (-4.05, 4.77)	-2.02 (-7.06, 3.01)
Fasting non-HDL-C, mg/dL	289	-4.65 (-8.24, -1.06)	146	-2.57 (-7.29, 2.15)	-2.23 (-7.69, 3.22)
Insulin resistance (HOMA-IR)	241	-0.13 (-0.69, 0.43)	124	0.47 (0.11, 0.84)	-0.29 (-0.98, 0.40)
eGFR from creatinine, mL/min/1.73m²	304	7.08 (5.77, 8.39)	155	-0.28 (-2.18, 1.63)	7.28 (5.02, 9.53)
eGFR from cystatin C, mL/min/1.73m²	298	-2.41 (-3.52, -1.30)	154	0.93 (-0.61, 2.47)	-3.54 (-5.35, -1.72)

[a]With 95% CIs based on the t-distribution. [b]With 95% CIs calculated from ANOVA models.
eGFR, estimated glomerular filtration rate; HOMA-IR, homeostatic model assessment for insulin resistance; n, number of participants with corresponding baseline and post-baseline data at Week 96.

Conclusions

- Switching to DOR/ISL (100/0.25 mg) from BIC/FTC/TAF maintained a high rate (89%) of virologic suppression and had similar efficacy to continuing BIC/FTC/TAF at Week 96
- DOR/ISL was generally well tolerated over 96 weeks; the AE profiles of DOR/ISL and BIC/FTC/TAF were comparable
- A switch to DOR/ISL had no adverse effects on total lymphocyte and CD4+ T-cell counts through Week 96

These findings are consistent with the study's primary Week 48 results.

Plain language summary

In this double-blind switch study, doravirine/islatravir (100/0.25 mg) taken once a day was similar to bictegravir/emtricitabine/tenofovir alafenamide taken once a day in maintaining low viral levels for 96 weeks. Doravirine/islatravir was generally well tolerated with a side effect profile comparable to bictegravir/emtricitabine/tenofovir alafenamide, and it did not negatively affect white blood cells.

References

1. Gibas KM, et al. *Lancet HIV.* 2022;9(12):e868-83.
2. Stockdale AJ, Khoo S. *Curr Opin HIV AIDS.* 2022;17(1):4-14.
3. Diamond TL, et al. *Antimicrob Agents Chemother.* 2022;66(6):e0013322.
4. Lai MT, et al. *Antimicrob Agents Chemother.* 2022; 66(5): e0222321.
5. Colson AE, et al. *Lancet* 2026; 407 (10528): 611-21.

Acknowledgments

We thank the study participants, their families, and all investigators and site personnel. We are also grateful for critical review of the data by scientific advisors.

Funding for this research was provided by Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA. Medical writing assistance was provided by Dominik Wolf, MSc, and editorial assistance was provided by Carol Zecca, BS, both of Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA.

Presented at American Conference for the Treatment of HIV (ACTHIV); April 30-May 2, 2026; Atlanta, GA, USA.

Copyright © 2026 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.