Pharmacokinetics and Safety of MK-8527 in Adults With Moderate or Severe Renal Impairment

Russ P. Carstens[1]; Yash Kapoor[1]; Arinjita Bhattacharyya[1]; Bhavna Kantesaria[1]; Graigory C. Garrett[1]; Gillian Gillespie[1]; Ryan Vargo[1]; S. Aubrey Stoch[1,†]; Helen Manthos[2]; Abe Lambert[2]; Jesse C. Nussbaum[1]; Boris Renjifo*

[1]Merck & Co., Inc., Rahway, NJ, USA; [2]Celerion, Lincoln, NE, USA

*Presenting on behalf of the authors

[†]An employee of Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA, at the time the study was conducted

Background

- Despite the approval of several agents for pre-exposure prophylaxis (PrEP), approximately 1.3 million people acquired HIV in 2024[1]
 - Long-acting oral options for HIV-1 PrEP, with longer dosing intervals, are needed
- MK-8527 is a novel, oral, nucleoside reverse transcriptase translocation inhibitor (NRTTI) in clinical development for HIV-1 prevention, with pharmacokinetic properties supporting once-monthly (QM) dosing, and is phosphorylated intracellularly to the pharmacologically active triphosphate (TP) form, MK-8527-TP[2-5]
 - In adults living with HIV-1 who had not previously taken antiretroviral agents, single doses of MK-8527 as low as 0.5 mg achieved a mean decrease in HIV-1 RNA of ≥1.0 $\log_{10}$ copies/mL at 7 days after dose administration[4]
 - A randomized, double-blind, placebo-controlled, Phase 2 study in adults with low likelihood of HIV-1 exposure demonstrated that 6 consecutive doses of MK-8527 (3 mg, 6 mg, and 12 mg QM) were generally well tolerated, with pharmacokinetics (PK) of MK-8527 and MK-8527-TP being dose proportional and well correlated, supporting clinical development of MK-8527[5]
 - Two Phase 3 studies of oral MK-8527 QM as PrEP are planned: enrollment has begun for EXPrESSIVE-11, while enrollment for EXPrESSIVE-10 will start later this year[6,7]
- Preclinical and clinical studies indicate that renal excretion plays a prominent role in NRTTI elimination[8-10]
- Renal impairment (RI) may significantly impact the clearance and PK of MK-8527, potentially leading to elevated plasma drug levels. Dose adjustment strategies could help ensure that plasma MK-8527 concentrations and MK-8527-TP in peripheral blood mononuclear cells (PBMCs) stay within the therapeutic range

Objective

- To evaluate the PK and safety of MK-8527 and MK-8527-TP in participants with moderate or severe RI compared with healthy participants with normal renal function

Methods

Study design

- In this open-label, single-dose, Phase 1 study (protocol MK-8527-008; NCT06295796), participants with normal renal function, moderate RI, or severe RI received a single dose of MK-8527 6 mg (**Figure 1**)
 - Normal renal function: estimated glomerular filtration rate (eGFR) ≥90 mL/min, calculated using the 2021 Chronic Kidney Disease Epidemiology Collaboration (CKD-EPI) equation
 - Moderate RI: eGFR 30-59 mL/min
 - Severe RI: eGFR 15-29 mL/min
- Healthy control participants with normal renal function were matched to the combined mean age (±15 years), mean body mass index (BMI; ±3.5 kg/m²), and sex (±2) of participants with moderate and severe RI
- Eligibility criteria included:
 - 18 to 75 years of age
 - BMI ≥18.0 and ≤40.0 kg/m²
- Exclusion criteria included:
 - History of alcohol or drug abuse, cancer, failed renal transplant, or end-stage renal disease requiring dialysis
 - Current pregnancy or lactation
 - Positive test result for HIV, hepatitis B virus surface antigen, or hepatitis C virus antibodies

Figure 1. Study design for protocol MK-8527-008

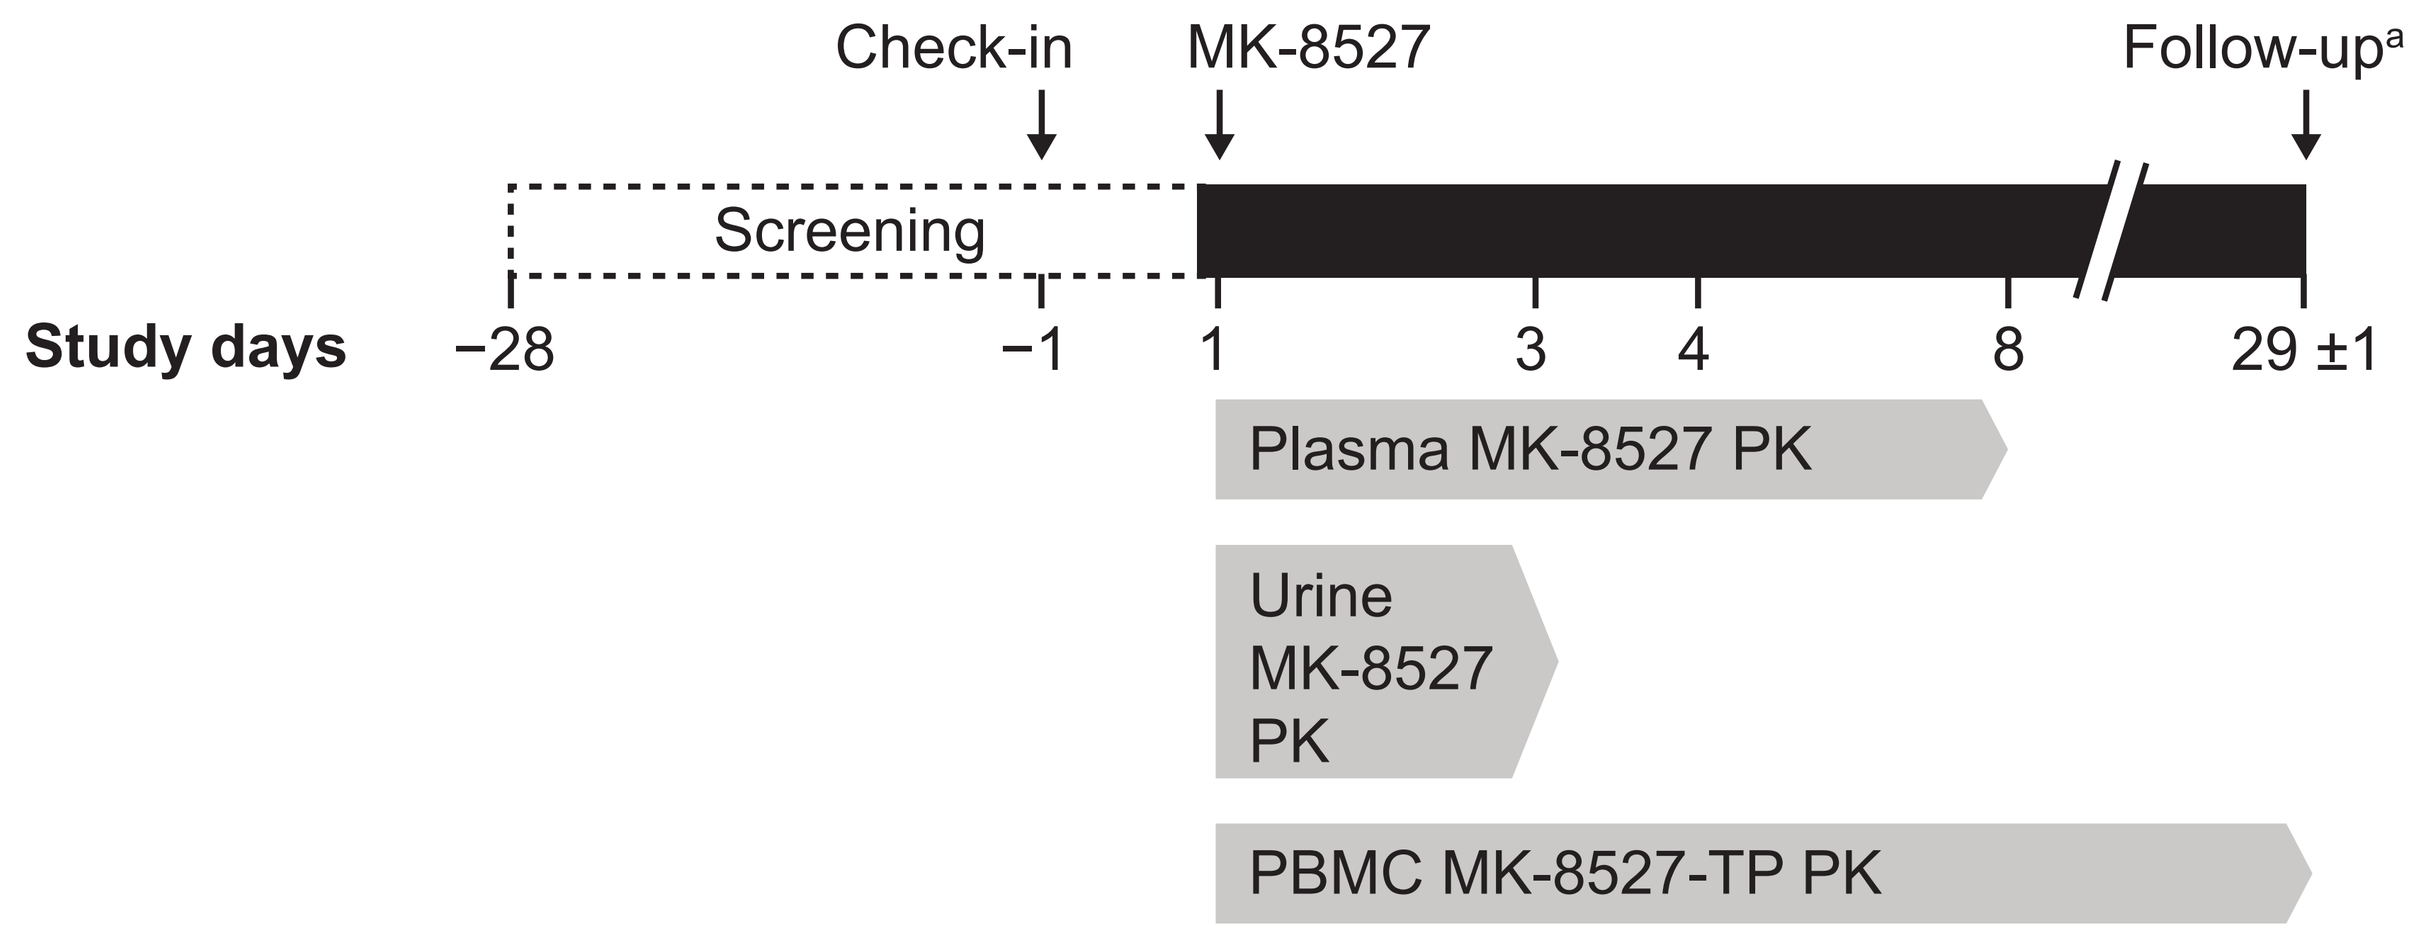
Check-in
MK-8527
Follow-up[a]
Screening
Study days
−28
−1
1
3
4
8
29 ±1
Plasma MK-8527 PK
Urine MK-8527 PK
PBMC MK-8527-TP PK

AE, adverse event; CRU, clinical research unit; MK-8527-TP, MK-8527 triphosphate; PBMC, peripheral blood mononuclear cell; PK, pharmacokinetics.
[a]All participants who received the study drug (including participants who terminated the study early) returned to the CRU on Day 29 (±1 day) for follow-up procedures and to determine if any AE had occurred since the last study visit.

Statistical analysis

- PK sampling was performed before dosing and up to 168 hours after dosing for plasma concentrations of MK-8527 and before dosing and up to 672 hours after dosing for intracellular concentrations of MK-8527-TP in PBMCs. Urine was collected before dosing and up to 48 hours after dosing and was assessed for concentrations of MK-8527
 - PK parameters of plasma MK-8527 that were analyzed included area under the concentration versus time curve from time 0 to infinity after single dosing ($AUC_{0-\infty}$), area under the concentration versus time curve from time 0 to the time of the last quantifiable (above lower limit of quantification) sample (AUC_{0-last}), maximum observed drug concentration after the administration of a given dose (C_{max}), time to maximum observed drug concentration (T_{max}), apparent terminal half-life ($t_{1/2}$), apparent clearance (CL/F), and apparent volume of distribution during terminal phase (Vz/F) of plasma MK-8527
 - PK parameters of intracellular MK-8527-TP in PBMCs that were analyzed included $AUC_{0-\infty}$, AUC_{0-last}, C_{max}, drug concentration at 168 hours (C_{168}), drug concentration at 672 hours (C_{672}), T_{max}, and $t_{1/2}$
 - Safety and tolerability were assessed through adverse event reporting and evaluation of vital signs, 12-lead electrocardiograms, and laboratory safety tests

Results

Study population

- Six participants were enrolled in each group (healthy control, moderate RI, severe RI; **Table 1**); all participants completed the study

Table 1. Participant demographics and baseline characteristics

Characteristic	Healthy control N = 6	Moderate RI N = 6	Severe RI N = 6
Age, median (range), years	60.0 (52-68)	65.0 (52-69)	70.0 (60-73)
Female, n (%)	3 (50)	2 (33)	3 (50)
Race, n (%)			
Black/African American	3 (50)	4 (67)	5 (83)
White	3 (50)	2 (33)	1 (17)
Ethnicity, n (%)			
Not Hispanic or Latino/a	5 (83)	6 (100)	6 (100)
Weight, median (range), kg	88.9 (78.8-100.4)	100.3 (86.2-116.3)	85.4 (64.6-105.5)
BMI, median (range), kg/m²	30.6 (28.7-33.7)	33.7 (30.5-37.5)	27.8 (25.3-34.3)
eGFR,[a] mean (SD), mL/min	100.8 (10.0)	44.3 (8.8)	22.9 (5.1)

BMI, body mass index; eGFR, estimated glomerular filtration rate; RI, renal impairment.
[a]Baseline eGFR (calculated using the 2021 Chronic Kidney Disease Epidemiology Collaboration equation) results were obtained twice (≥72 hours apart) during participant screening, and mean values were used to determine renal function. Values represent deindexed eGFR using the body surface area for each participant.

PK analysis of MK-8527

- Participants with moderate or severe RI had 1.79-fold or 2.24-fold higher plasma MK-8527 $AUC0_{-\infty}$, respectively, compared with healthy control participants (**Table 2; Figure 2**)
- Plasma MK-8527 C_{max} was generally similar between participants with moderate RI or severe RI and healthy control participants
- Participants with moderate or severe RI had 2.00-fold or 2.35-fold higher, respectively, intracellular MK-8527-TP $AUC_{0-\infty}$ and 2.16-fold or 1.79-fold higher intracellular MK-8527-TP C_{max}, respectively, compared with healthy control participants
- Participants with moderate or severe RI had longer $t_{1/2}$ for plasma MK-8527 and PBMC MK-8527-TP and decreased CL/F and Vz/F for plasma MK-8527 compared with healthy control participants (**Table 2**; **Figure 3**)
- Following the administration of a single oral dose of MK-8527 6 mg, the amount of unchanged MK-8527 recovered in urine was lower in both moderate and severe RI groups compared with healthy control participants, and appeared to decrease with increasing severity of RI

Table 2. Summary of PK parameter values of plasma MK-8527 and PBMC MK-8527-TP in participants with moderate RI or severe RI compared with matched healthy controls

Parameter	Geometric mean (% GCV)			GMR (90% CI)	
	Healthy control n = 6	Moderate RI n = 6	Severe RI n = 6	Moderate RI vs healthy control	Severe RI vs healthy control
Plasma MK-8527					
C_{max}, nM	112 (25.7)	161 (48.9)	91.7 (22.1)	1.43 (0.96-2.14)	0.82 (0.64-1.05)
T_{max},[a] h	0.75 (0.52, 1.53)	0.75 (0.50, 1.00)	0.99 (0.25, 6.00)	NA	NA
$AUC_{0-\infty}$, nM·h	918 (22.2)	1640 (30.0)	2060 (24.0)	1.79 (1.36-2.35)	2.24 (1.77-2.85)
AUC_{0-last}, nM·h	769 (21.4)	1160 (24.6)	1430 (20.1)	1.51 (1.19-1.92)	1.86 (1.50-2.31)
$t_{1/2}$, h	84.2 (14.1)	118 (32.5)	113 (38.0)	NA	NA
CL/F, L/h	21.2 (22.2)	11.8 (30.0)	9.45 (24.0)	NA	NA
Vz/F, L	2570 (29.7)	2020 (27.0)	1540 (36.5)	NA	NA
PBMC MK-8527-TP					
C_{max}, pmol/10^6 cells	1.77 (28.4)	3.82 (78.4)	3.18 (14.7)	2.16 (1.20-3.86)	1.79 (1.41-2.28)
T_{max},[a] h	23.08 (11.92, 23.92)	23.92 (23.53, 167.07)	23.01 (23.00, 23.53)	NA	NA
$AUC_{0-\infty}$, h·pmol/10^6 cells	235 (35.9)	468 (27.5)	552 (5.9)	2.00 (1.42-2.81)	2.35 (1.76-3.14)
AUC_{0-last}, h·pmol/10^6 cells	217 (37.2)	493 (51.6)	588 (37.6)	2.26 (1.44-3.56)	2.71 (1.85-3.95)
$t_{1/2}$, h	170 (44.1)	212 (24.6)	251 (36.2)	NA	NA

$AUC_{0-\infty}$, area under the concentration-time curve from time 0 to infinity; AUC_{0-last}, area under the concentration-time curve from time 0 to last sampling time after dose; CL/F, apparent clearance; C_{max}, maximum concentration; GCV, geometric coefficient of variation; GMR, geometric mean ratio; NA, not available; PBMC, peripheral blood mononuclear cell; PK, pharmacokinetics; RI, renal impairment; T_{max}, time to C_{max}; $t_{1/2}$, apparent terminal half-life; Vz/F, apparent volume of distribution during terminal phase.
[a]T_{max} is presented as median (range).

Figure 2. Mean plasma MK-8527 concentration over time

(A) and individual $AUC_{0-\infty}$ and geometric mean (95% CI). (B) in participants with moderate and severe RI and matched healthy controls.

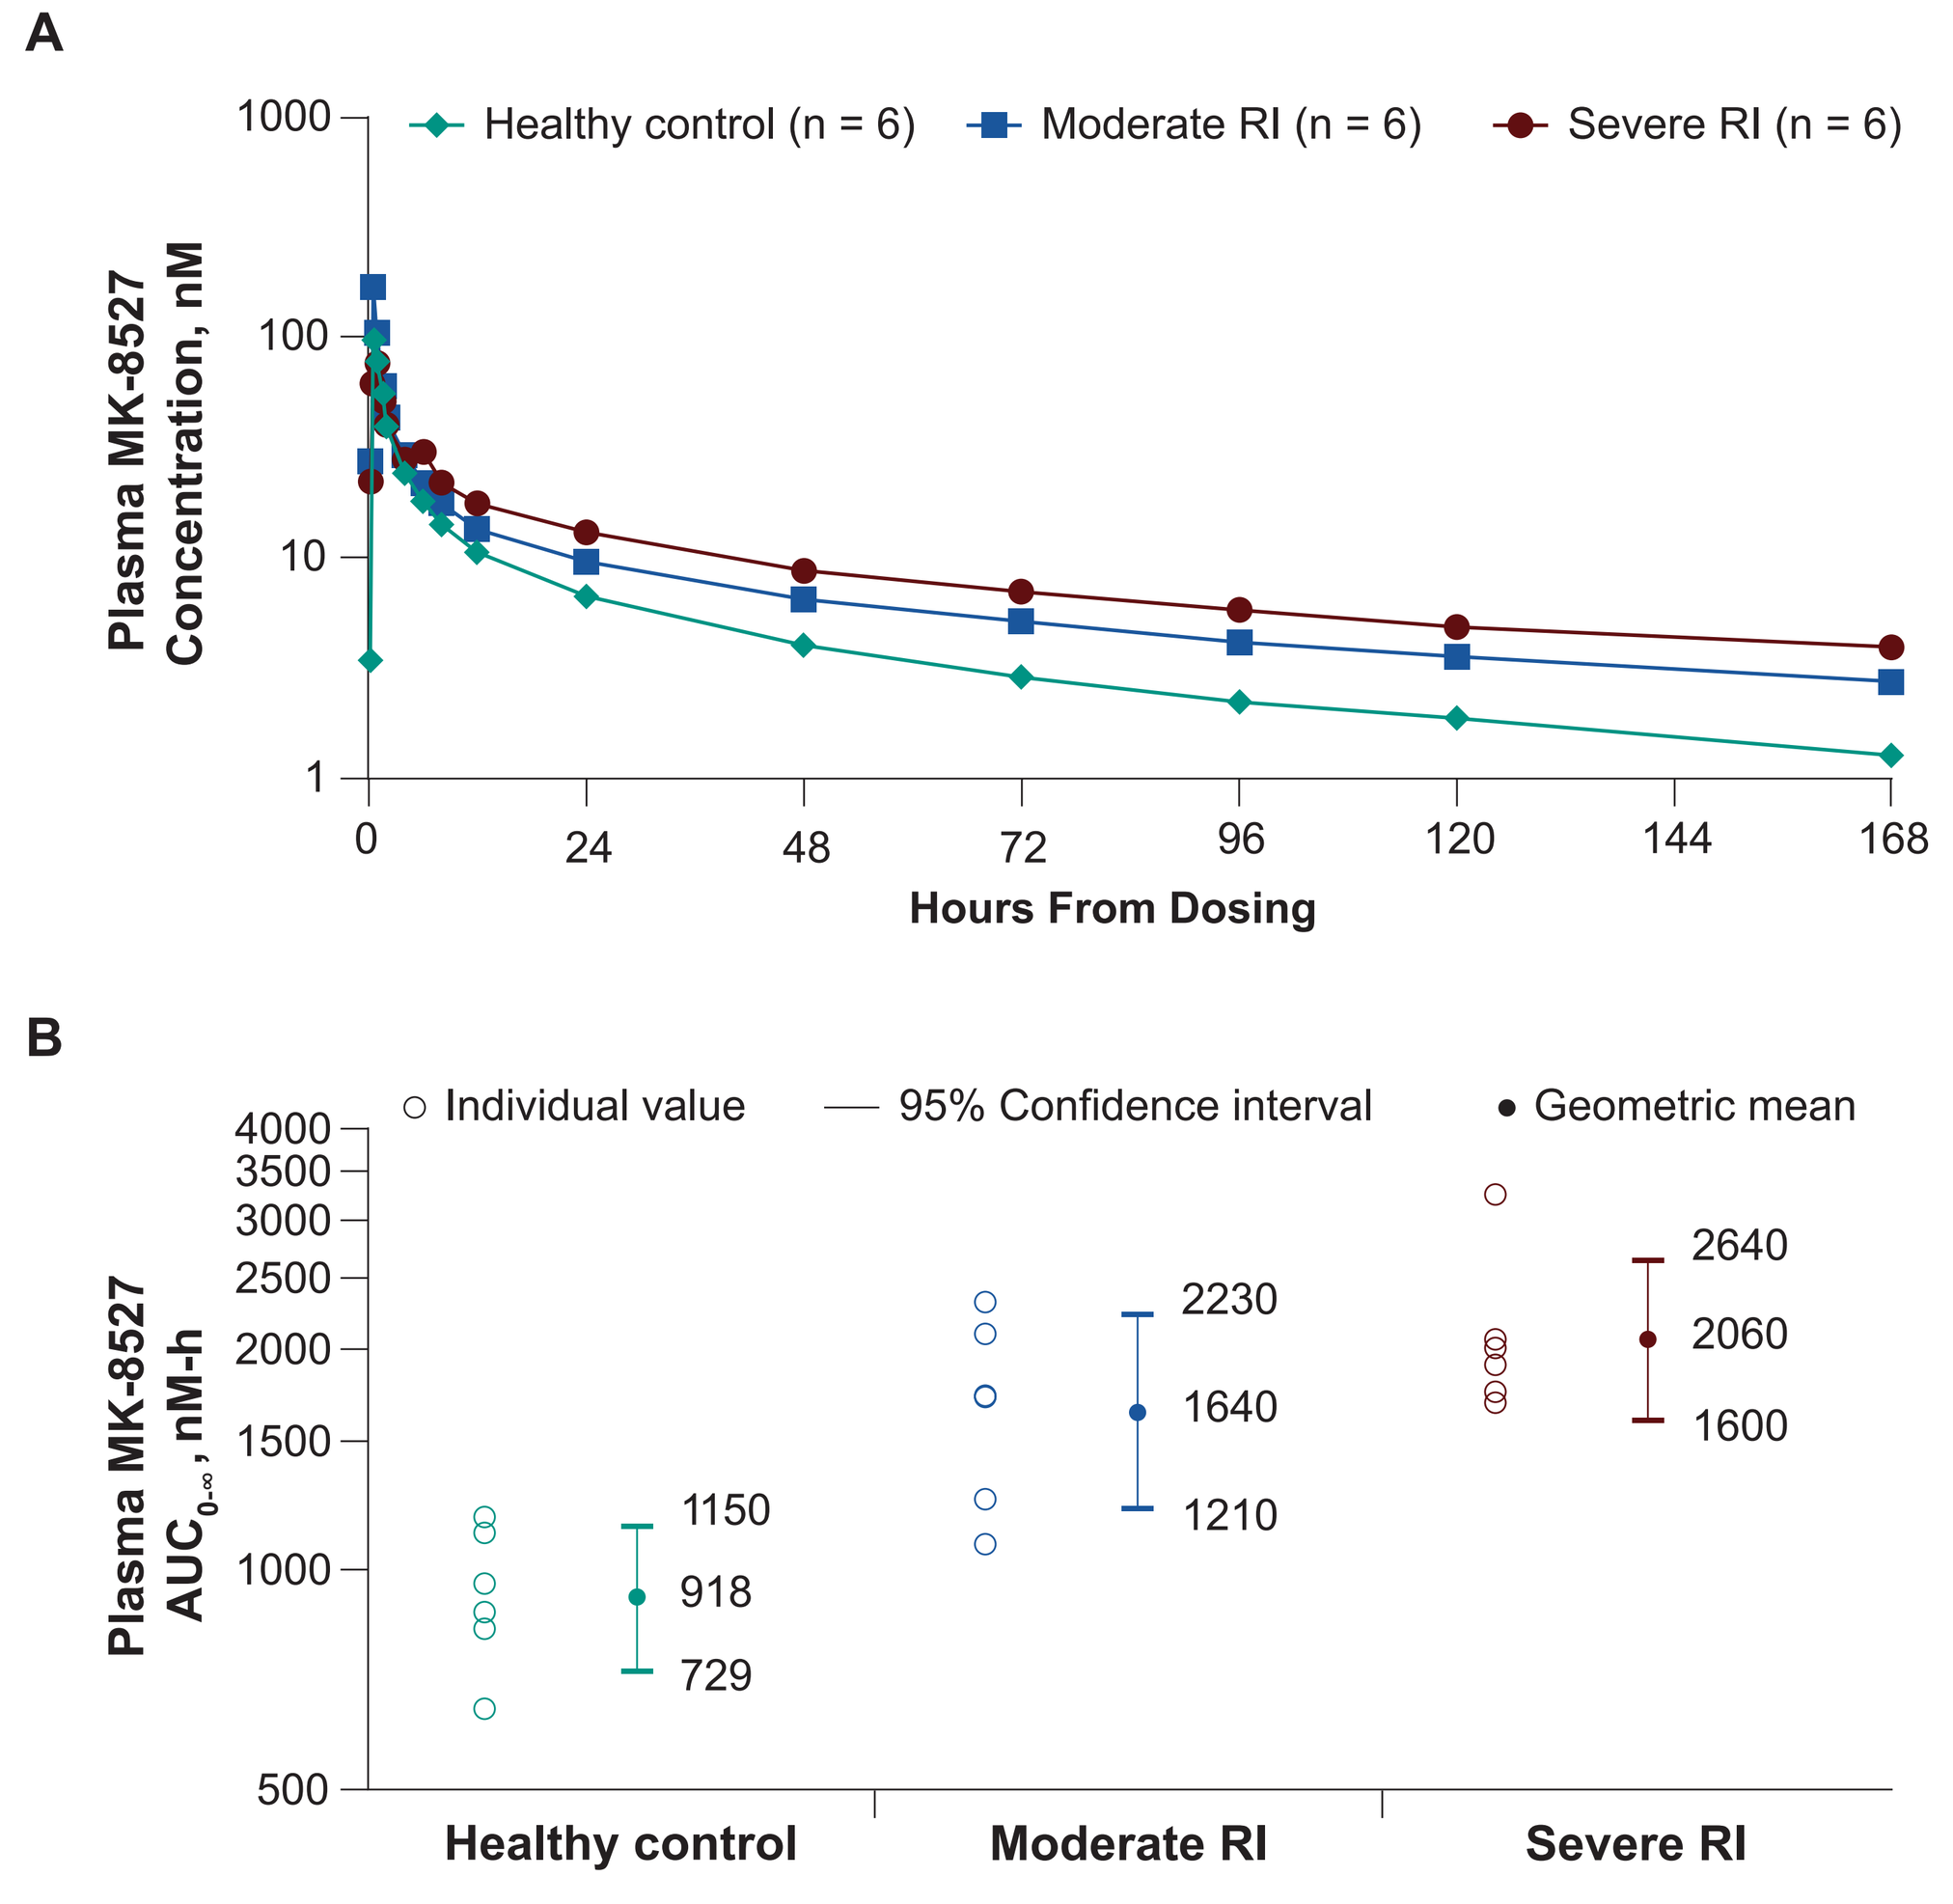
A
Healthy control (n = 6)
Moderate RI (n = 6)
Severe RI (n = 6)
Plasma MK-8527 Concentration, nM
1000
100
10
1
0
24
48
72
96
120
144
168
Hours From Dosing
B
Individual value
95% Confidence interval
Geometric mean
Plasma MK-8527 AUC0-∞, nM·h
4000
3500
3000
2500
2000
1500
1000
500
1150
918
729
2230
1640
1210
2640
2060
1600
Healthy control
Moderate RI
Severe RI

$AUC_{0-\infty}$, area under the concentration-time curve from time 0 to infinity; RI, renal impairment.

Figure 3. Mean PBMC MK-8527-TP concentration over time

(A) and individual $AUC_{0-\infty}$ and geometric mean (95% CI). (B) in participants with moderate and severe RI and matched healthy controls.

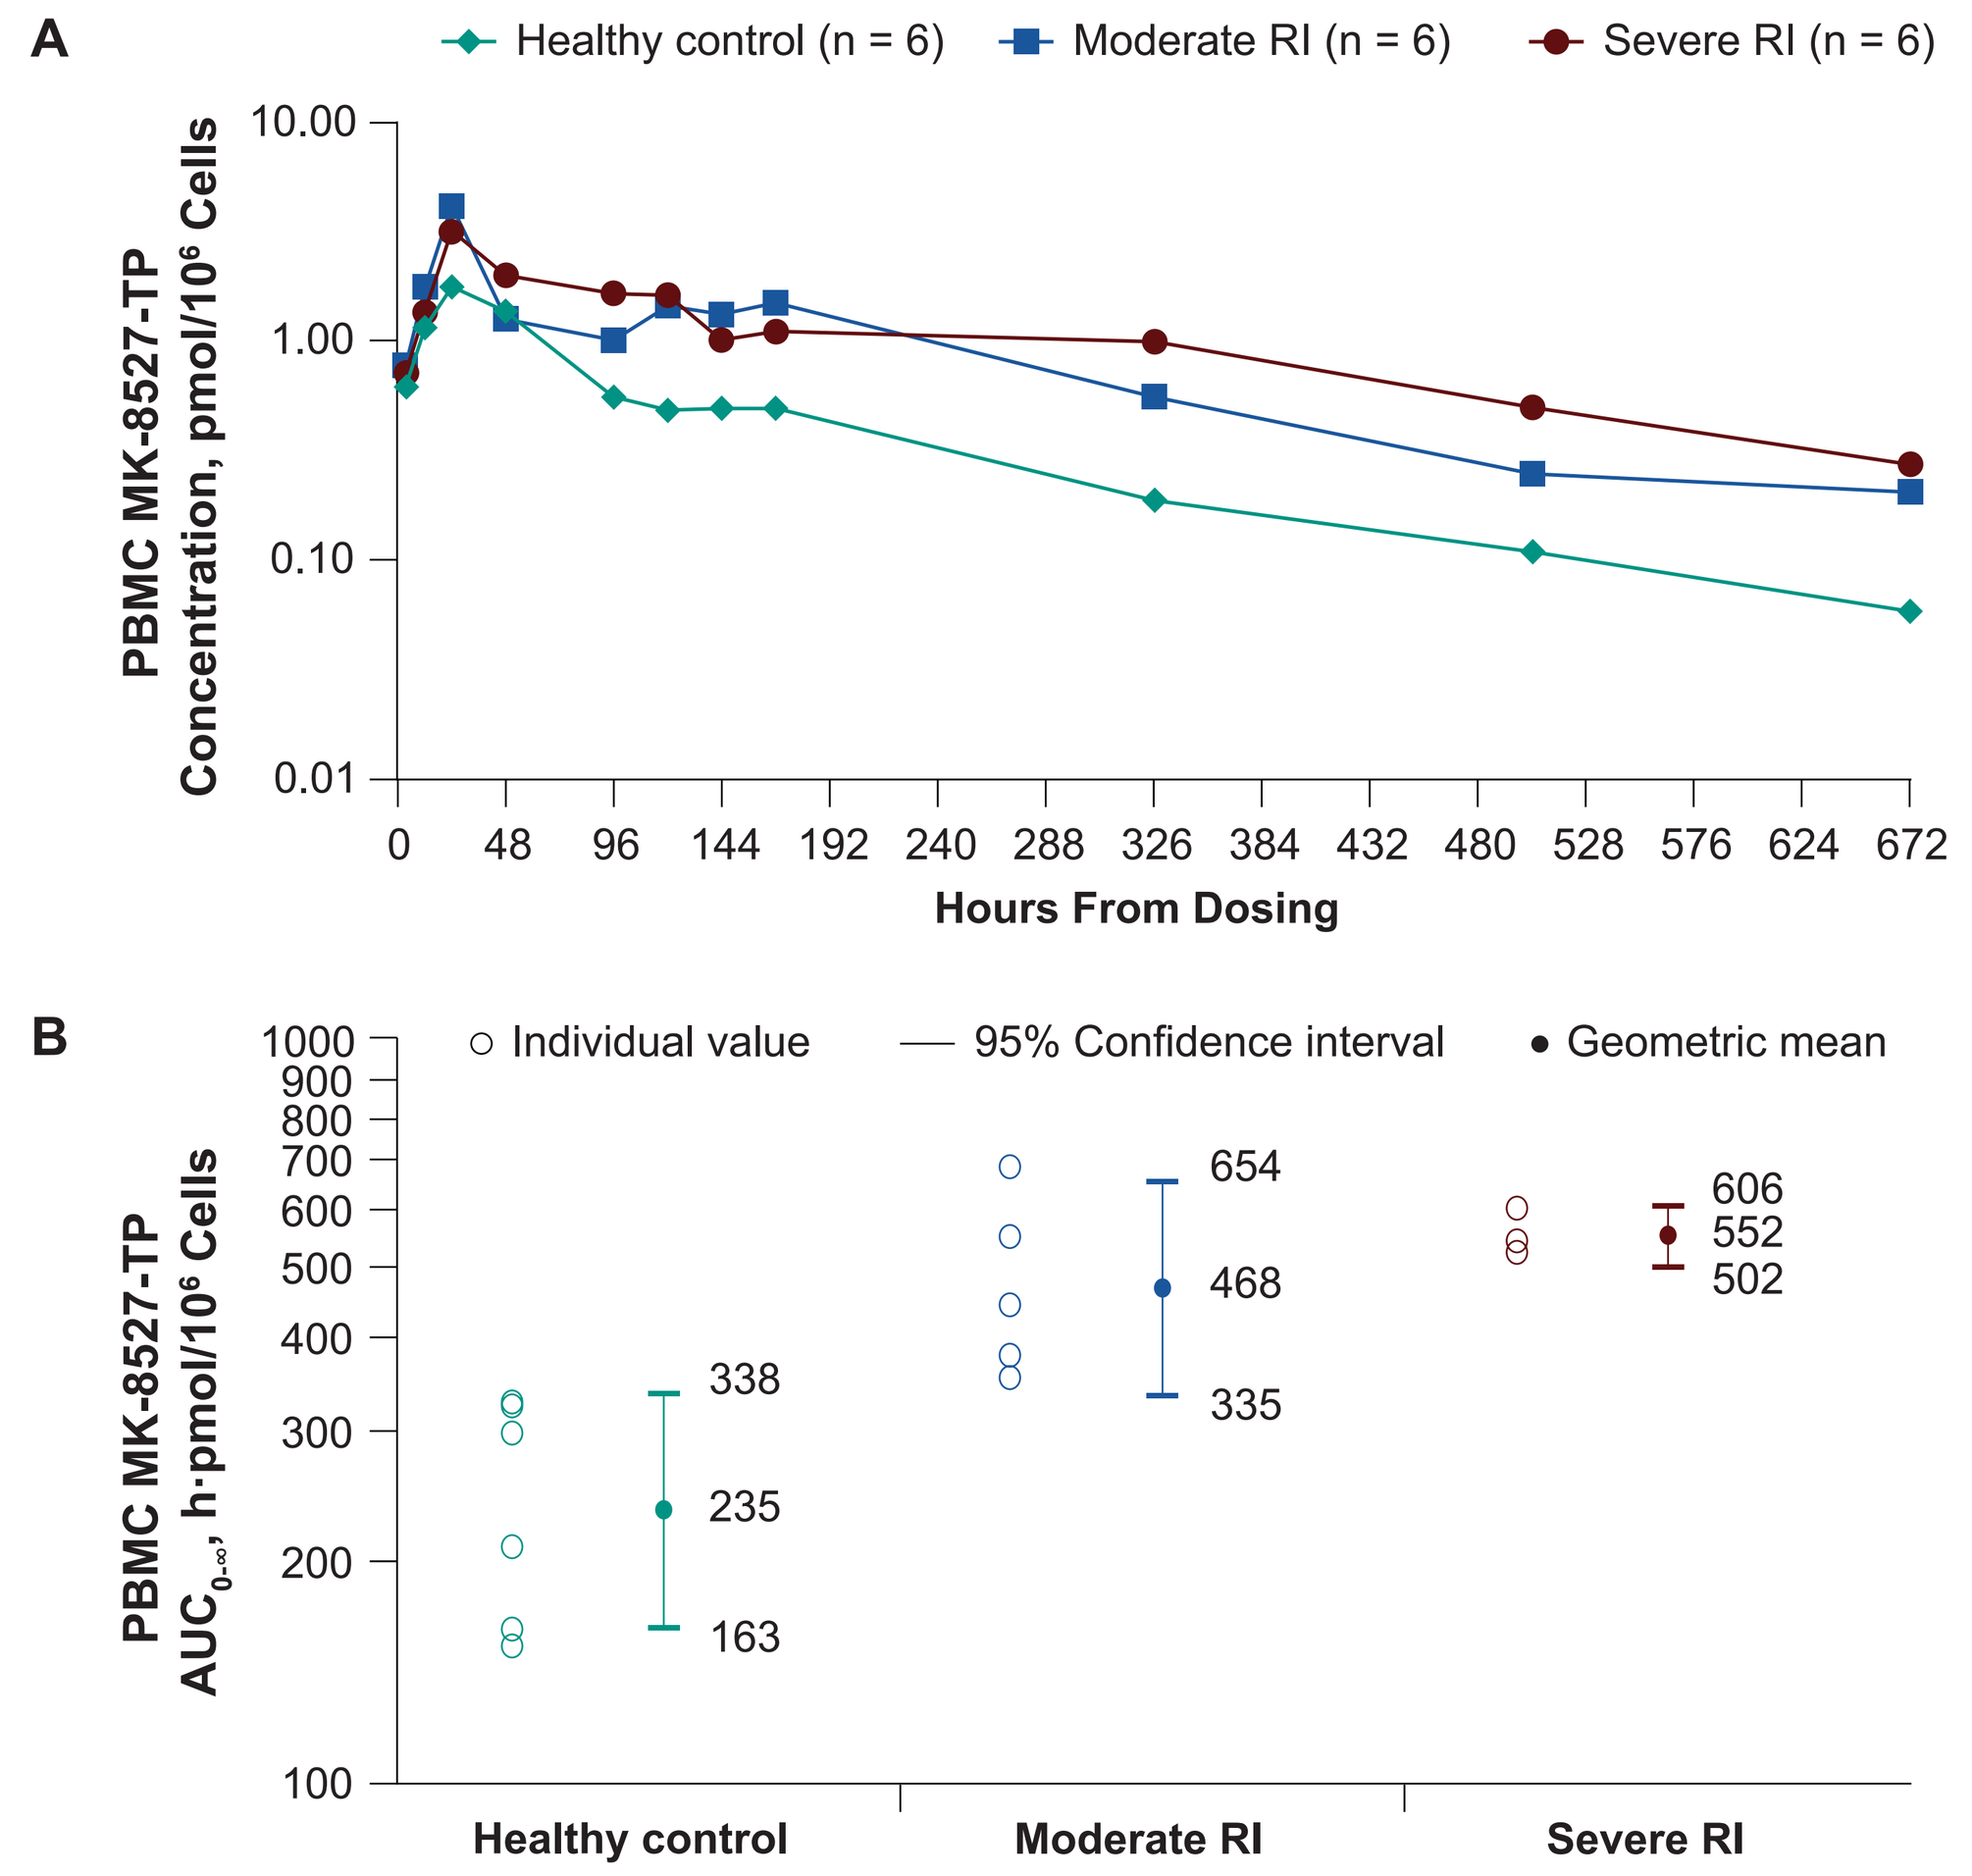
A
Healthy control (n = 6)
Moderate RI (n = 6)
Severe RI (n = 6)
PBMC MK-8527-TP Concentration, pmol/10⁶ Cells
10.00
1.00
0.10
0.01
0
48
96
144
192
240
288
326
384
432
480
528
576
624
672
Hours From Dosing
B
Individual value
95% Confidence interval
Geometric mean
PBMC MK-8527-TP AUC0-∞, h·pmol/10⁶ Cells
1000
900
800
700
600
500
400
300
200
100
338
235
163
654
468
335
606
552
502
Healthy control
Moderate RI
Severe RI

$AUC0_{-\infty}$, area under the concentration-time curve from time 0 to infinity; PBMC, peripheral blood mononuclear cell; RI, renal impairment; TP, triphosphate.

Safety

- No adverse events were reported in this study
- There were no significant findings concerning vital signs, laboratory safety tests, and electrocardiogram results after MK-8527 treatment

Conclusions

- Consistent with renal excretion being a primary pathway for MK-8527 elimination, plasma MK-8527 exposure was higher in participants with moderate or severe RI than in healthy control participants with normal renal function
- Exposure of intracellular MK-8527-TP was higher in participants with moderate or severe RI than in healthy control participants with normal renal function
- A single dose of MK-8527 was well tolerated in participants with moderate or severe RI

References

1. UNAIDS Fact Sheet 2025 - Global HIV Statistics. Accessed August 28, 2025. https://www.unaids.org/sites/default/files/media_asset/UNAIDS_FactSheet_en.pdf
2. Raheem IT, et al. MK-8527 is a novel inhibitor of HIV-1 reverse transcriptase translocation with potential for extended-duration dosing. *PLoS Biol.* Aug 2025;23(8):e3003308.
3. Gillespie G, et al. Safety and pharmacokinetics of MK-8527, a novel NRTTI, in adults without HIV. Presented at: Conference on Retroviruses and Opportunistic Infections (CROI); March 3-6, 2024; Denver, CO, USA.
4. Carstens R, et al. Single dose administration of MK-8527, a novel NRTTI, in adults with HIV-1. Presented at: Conference on Retroviruses and Opportunistic Infections (CROI); March 3-9, 2024; Denver, CO, USA.
5. Mayer K, et al. Safety and pharmacokinetics of MK-8527 oral once-monthly: a phase 2 study in adults at low risk of HIV-1 exposure. Presented at: International AIDS Society (IAS) Conference; July 13-17, 2025; Kigali, Rwanda.
6. ClinicalTrials.gov. NCT07071623. A study of MK-8527 to prevent human immunodeficiency virus type 1 (HIV-1) (MK-8527-010). Accessed August 28, 2025. https://www.clinicaltrials.gov/study/NCT07071623
7. ClinicalTrials.gov. NCT07044297. A clinical study of MK-8527 to prevent human immunodeficiency virus type 1 (HIV-1) (MK-8527-011). Accessed August 28, 2025. https://www.clinicaltrials.gov/study/NCT07044297
8. Bleasby K, et al. Islatravir is not expected to be a victim or perpetrator of drug-drug interactions via major drug-metabolizing enzymes or transporters. *Viruses.* 2021;13(8):1566.
9. Matthews RP, et al. Safety and pharmacokinetics of islatravir in individuals with severe renal insufficiency. *Antimicrob Agents Chemother.* 2022;66(12):e0093122.
10. Fillgrove KL, et al. Nonclinical and clinical characterization of the absorption, metabolism, and excretion of islatravir. *Antimicrob Agents Chemother.* 2025;69(2):e0103024.

Acknowledgments

The authors thank all the individuals who participated in this study. The contributions of the investigators and their staff are also gratefully recognized. Medical writing and/or editorial assistance was provided by Agnieszka Looney, PhD, Viola Kooij, PhD, and Jared Cochran, PhD, of ApotheCom (Yardley, PA, USA). This assistance was funded by Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA. Funding for this research was provided by Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA.

Contact information

Contact the author jesse.nussbaum@merck.com

Presented at American Conference for the Treatment of HIV (ACTHIV); April 30-May 2, 2026; Atlanta, GA, USA.

Copyright © 2026 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All rights reserved.