ARTISTRY-2
58

Phase 3 Efficacy and Safety of Switch From B/F/TAF to Single-Tablet BIC/LEN in ARTISTRY-2

Eric G Meissner[1], Moti Ramgopal[2], Peter J Ruane[3], William Sanchez[4], Jean-Pierre Routy[5], Mark O'Reilly[6], Sergio Lupo[7], Sven Schellberg[8], Keith Aizen[9], Hailin Huang[9], Kwad Mponponsuo[9], Peter Sklar[9], Henry Nguyen[9*], Rachel Rogers [9*], Martin Rhee[9], Chien-Ching Hung[10]

[1]Medical University of South Carolina, Charleston, SC, USA; [2]Midway Immunology and Research Center, Fort Pierce, FL, USA; [3]Ruane Clinical Research Group, Los Angeles, CA, USA; [4]Floridian Clinical Research, Miami Lakes, FL, USA; [5]McGill University Health Centre, Montreal, QC, Canada; [6]Taylor Square Private Clinic, Sydney, NSW, Australia; [7]CAICI, Rosario, Argentina; [8]Novopraxis Berlin GbR, Berlin, Germany; [9]Gilead Sciences, Inc., Foster City, CA, USA; [10]National Taiwan University Hospital Yunlin Branch, Yunlin, Taiwan

*Listed as author for presentation purposes only with permission of all authors

Conclusions

- A novel single-tablet regimen (STR) of bictegravir/lenacapavir (BIC/LEN) maintained high levels of virologic suppression at Week 48 and was noninferior to standard-of-care treatment with bictegravir/emtricitabine/tenofovir alafenamide (B/F/TAF) in people with HIV with virologic suppression
- BIC/LEN was generally well tolerated
 - There were no serious drug-related adverse events (AEs)
 - Grade 3 or higher drug-related AEs were infrequent
 - AEs leading to discontinuation were infrequent
- These results are consistent with those of ARTISTRY-1[1] and support the use of a novel BIC/LEN STR to expand treatment options for people with HIV with virologic suppression

For primary outcomes from Phase 3 of ARTISTRY-1, see Oral 181 "Phase 3 Efficacy and Safety of Switch from Complex Regimen to Single-Tablet BIC/LEN in ARTISTRY-1" presented by Prof. Chloe Orkin (February 25, 2026; 10 a.m. – 12 p.m.)

Plain Language Summary

- Bictegravir and lenacapavir (BIC/LEN) is being studied as a new single-tablet treatment for human immunodeficiency virus (HIV) that is taken as one pill every day
- In this study, BIC/LEN was similarly effective for controlling the amount of HIV in participants' blood and had similar rates of side effects as the commonly used HIV treatment, bictegravir/emtricitabine/tenofovir alafenamide (B/F/TAF), over 48 weeks

Introduction

STRs have transformed the HIV treatment landscape and are associated with significant benefits, including improved clinical outcomes, adherence, and quality of life, for people with HIV[2,3]

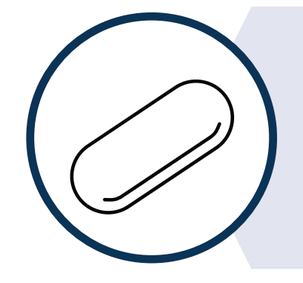

A novel BIC/LEN STR could expand treatment options for people with HIV with virologic suppression

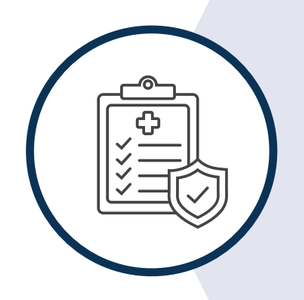

ARTISTRY-1 demonstrated that a once-daily BIC/LEN STR was noninferior to complex regimens in maintaining high rates of virologic suppression through 48 weeks in adults with HIV with a high prevalence of comorbidities, polypharmacy, and prior viral resistance[1]

Objective

- We evaluated the efficacy and safety of a once-daily BIC/LEN STR in people with HIV with virologic suppression who were switching from B/F/TAF in the Phase 3 ARTISTRY-2 trial

Methods

- ARTISTRY-2 (NCT06333808) is a Phase 3, randomized, double-blind, active-controlled multicenter trial

Study Design of ARTISTRY-2

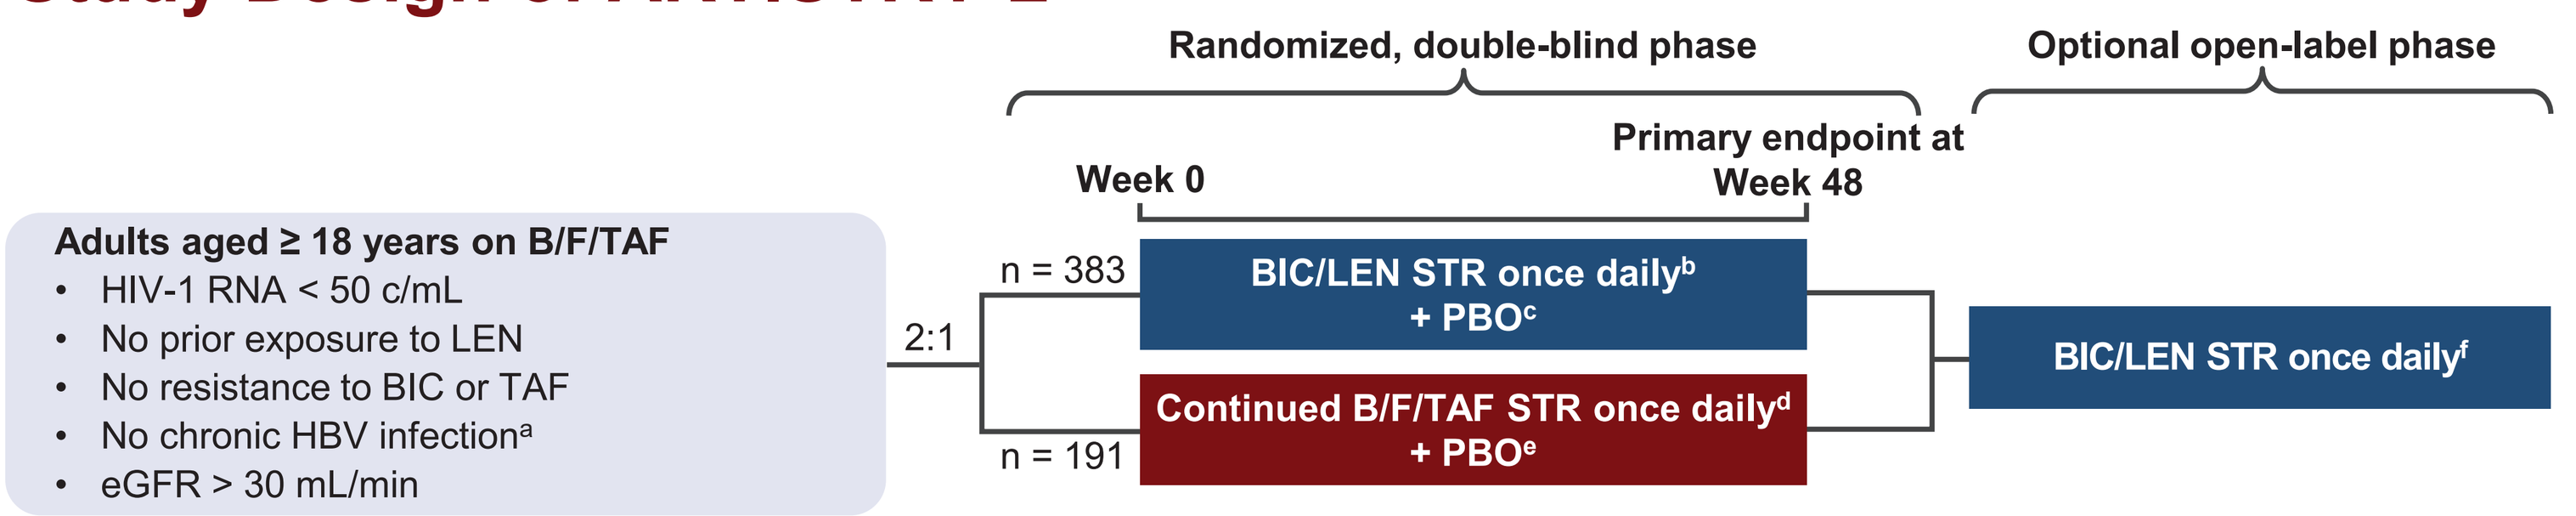
Randomized, double-blind phase
Optional open-label phase
Primary endpoint at Week 48
Week 0
Adults aged ≥ 18 years on B/F/TAF
HIV-1 RNA < 50 c/mL
No prior exposure to LEN
No resistance to BIC or TAF
No chronic HBV infection[a]
eGFR > 30 mL/min
2:1
n = 383
n = 191
BIC/LEN STR once daily[b] + PBO[c]
Continued B/F/TAF STR once daily[d] + PBO[e]
BIC/LEN STR once daily[f]

Endpoints
- **Primary:**
 - HIV-1 RNA ≥ 50 c/mL (US FDA Snapshot algorithm; 4% noninferiority margin) at Week 48
- **Secondary:**
 - HIV-1 RNA < 50 c/mL (US FDA Snapshot algorithm) at Week 48
 - Change from baseline in CD4 cell count at Week 48
 - Treatment-emergent AEs through Week 48

[a]Determined by the following at the screening visit: either positive HBV surface antigen and negative HBV surface antibody regardless of HBV core antibody status, or positive HBV core antibody and negative HBV surface antibody regardless of HBV surface antigen status. [b]BIC/LEN 75/50 mg; participants received an oral loading dose of LEN 600 mg on Days 1 and 2 of treatment. [c]Placebo-to-match B/F/TAF. [d]B/F/TAF 50/200/25 mg; participants received placebo-to-match LEN on Days 1 and 2 of treatment. [e]Placebo-to-match BIC/LEN. [f]BIC/LEN 75/50 mg.
AE, adverse event; BIC, bictegravir; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; c, copies; CD4, cluster of differentiation 4; eGFR, estimated glomerular filtration rate; FDA, Food and Drug Administration; HBV, hepatitis B virus; LEN, lenacapavir; PBO, placebo; STR, single-tablet regimen; TAF, tenofovir alafenamide.

Results

Baseline Demographic and Disease Characteristics

	BIC/LEN n = 383	B/F/TAF n = 191	Total N = 574
Age, years, median (range)	47 (23-77)	51 (26-77)	49 (23-77)
≥ 55 years, n (%)	135 (35.2)	70 (36.6)	205 (35.7)
Assigned female at birth, n (%)	69 (18.0)	42 (22.0)	111 (19.3)
Race, n (%)			
White	199 (52.0)	111 (58.1)	310 (54.0)
Black	113 (29.5)	44 (23.0)	157 (27.4)
Asian	51 (13.3)	26 (13.6)	77 (13.4)
Other[a]	20 (5.2)	10 (5.2)	30 (5.2)
Hispanic or Latine,[b] n (%)	106 (27.7)	49 (25.8)	155 (27.1)
CD4 count, cells/μL, median (Q1, Q3)	711 (547, 914)	663 (522, 873)	695 (541, 902)
Selected comorbidities,[c,d] n (%)			
Dyslipidemia	158 (41.3)	79 (41.4)	237 (41.3)
Hypertension	141 (36.8)	63 (33.0)	204 (35.5)
Hyperglycemia/diabetes mellitus	68 (17.8)	31 (16.2)	99 (17.2)
Chronic kidney disease	20 (5.2)	12 (6.3)	32 (5.6)
Number of selected comorbidities, n (%)			
1	94 (24.5)	57 (29.8)	151 (26.3)
≥ 2	121 (31.6)	52 (27.2)	173 (30.1)

[a]Category includes American Indian or Alaska Native, Native Hawaiian, Pacific Islander, and other. [b]Local regulators did not allow the collection of ethnicity information for one participant in the B/F/TAF group; percentage was calculated using participants with available information as the denominator. [c]Categories are not mutually exclusive. [d]Grouped terms on standardized MedDRA query narrow search. BIC, bictegravir; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; CD4, cluster of differentiation 4; LEN, lenacapavir; MedDRA, Medical Dictionary for Regulatory Activities; Q, quartile.

Virologic Outcomes at Week 48 (US FDA Snapshot Analysis)

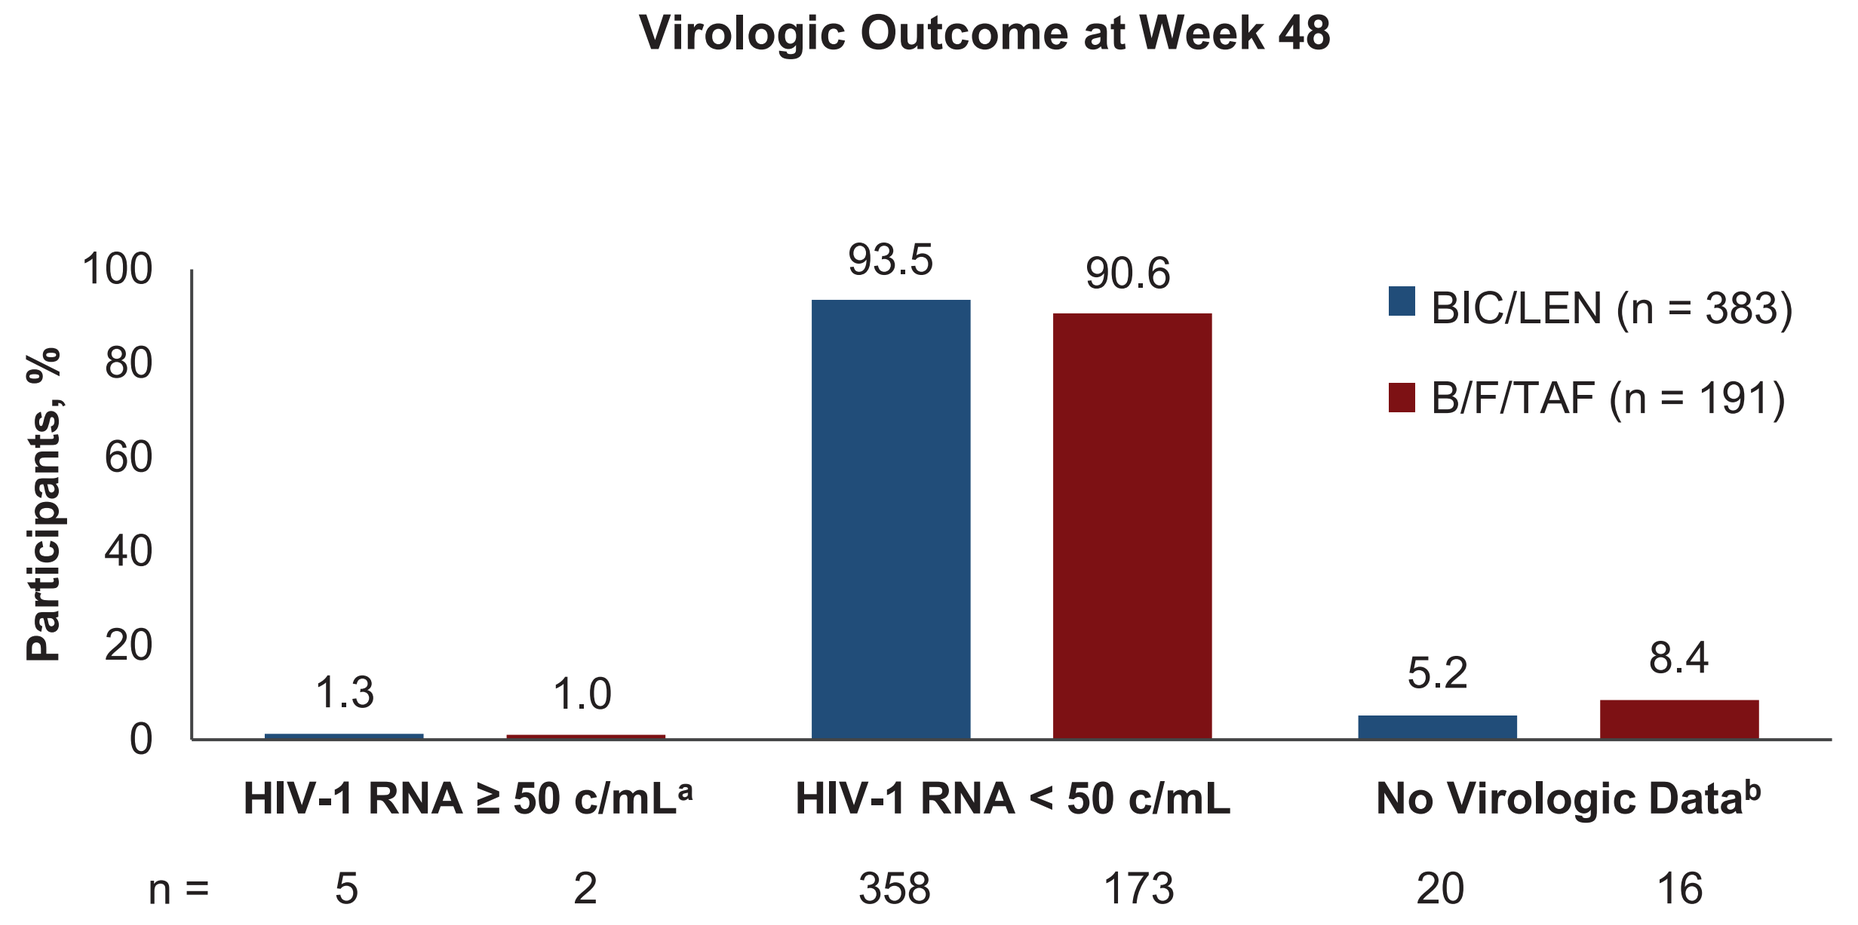
Virologic Outcome at Week 48
Participants, %
100
80
60
40
20
0
1.3
1.0
93.5
90.6
5.2
8.4
BIC/LEN (n = 383)
B/F/TAF (n = 191)
HIV-1 RNA ≥ 50 c/mL[a]
HIV-1 RNA < 50 c/mL
No Virologic Data[b]
n =
5
2
358
173
20
16

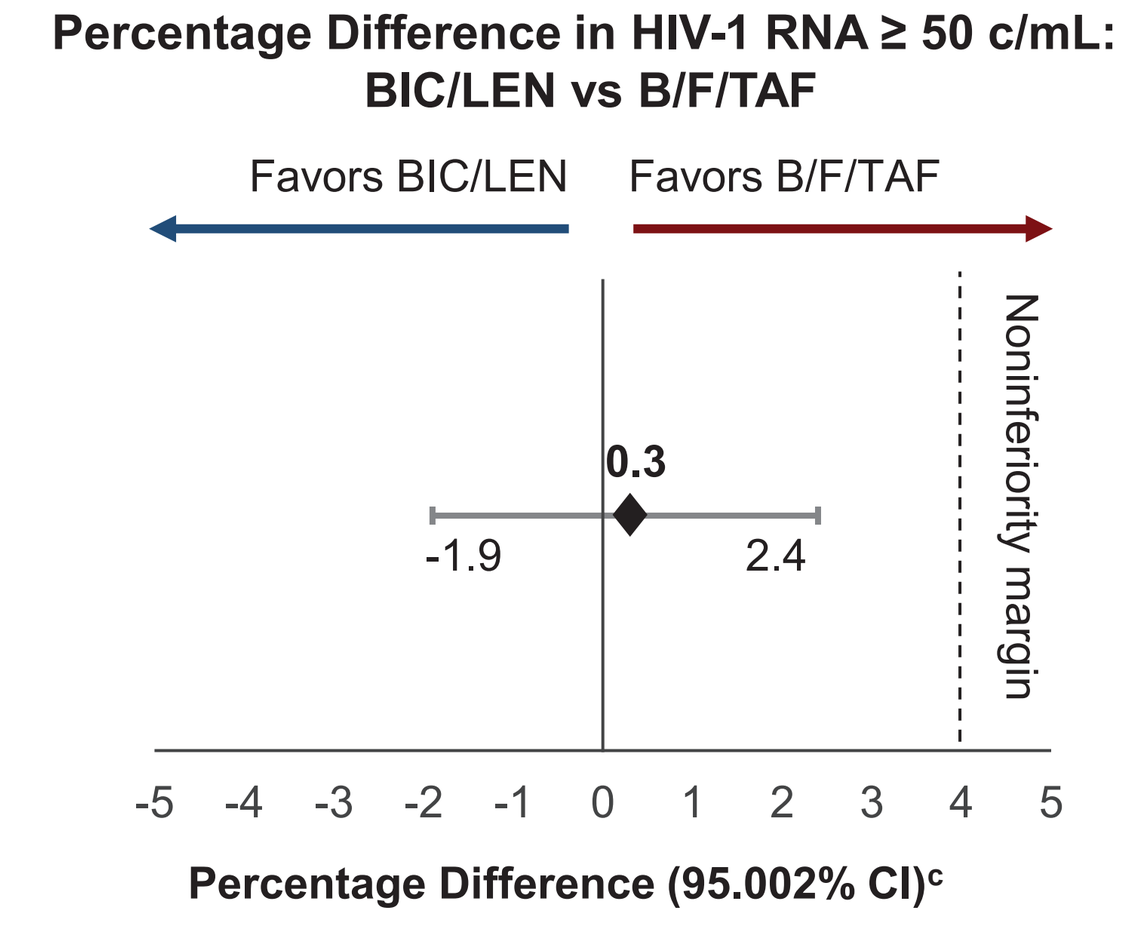
Percentage Difference in HIV-1 RNA ≥ 50 c/mL: BIC/LEN vs B/F/TAF
Favors BIC/LEN
Favors B/F/TAF
0.3
-1.9
2.4
Noninferiority margin
-5 -4 -3 -2 -1 0 1 2 3 4 5
Percentage Difference (95.002% CI)[c]

[a]Discontinued study drug due to other reasons and last available HIV-1 RNA was ≥ 50 c/mL: BIC/LEN n = 3, B/F/TAF n = 2. [b]Discontinued study drug due to adverse event or death: BIC/LEN n = 6, B/F/TAF n = 4; discontinued study drug due to other reasons and last available HIV-1 RNA was < 50 c/mL: BIC/LEN n = 14, B/F/TAF n = 12. [c]Difference in percentages of participants between treatment groups (BIC/LEN minus B/F/TAF) and two-sided CIs were constructed based on Mantel-Haenszel stratum weights and Koch variance estimator and adjusted by geographic region (US vs non-US).
BIC, bictegravir; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; c, copies; FDA, Food and Drug Administration; LEN, lenacapavir.

- In the BIC/LEN group, four of five participants with HIV-1 RNA ≥ 50 copies (c)/mL at Week 48 had low-level viremia (HIV-1 RNA < 1000 c/mL)
- One participant in the BIC/LEN group had HIV-1 RNA 10,700 c/mL at Week 48 and resuppressed following change to a protease inhibitor–containing regimen

Once-daily oral BIC/LEN STR was noninferior to standard-of-care treatment with B/F/TAF in maintaining virologic suppression at 48 weeks

CD4 Count Over Time

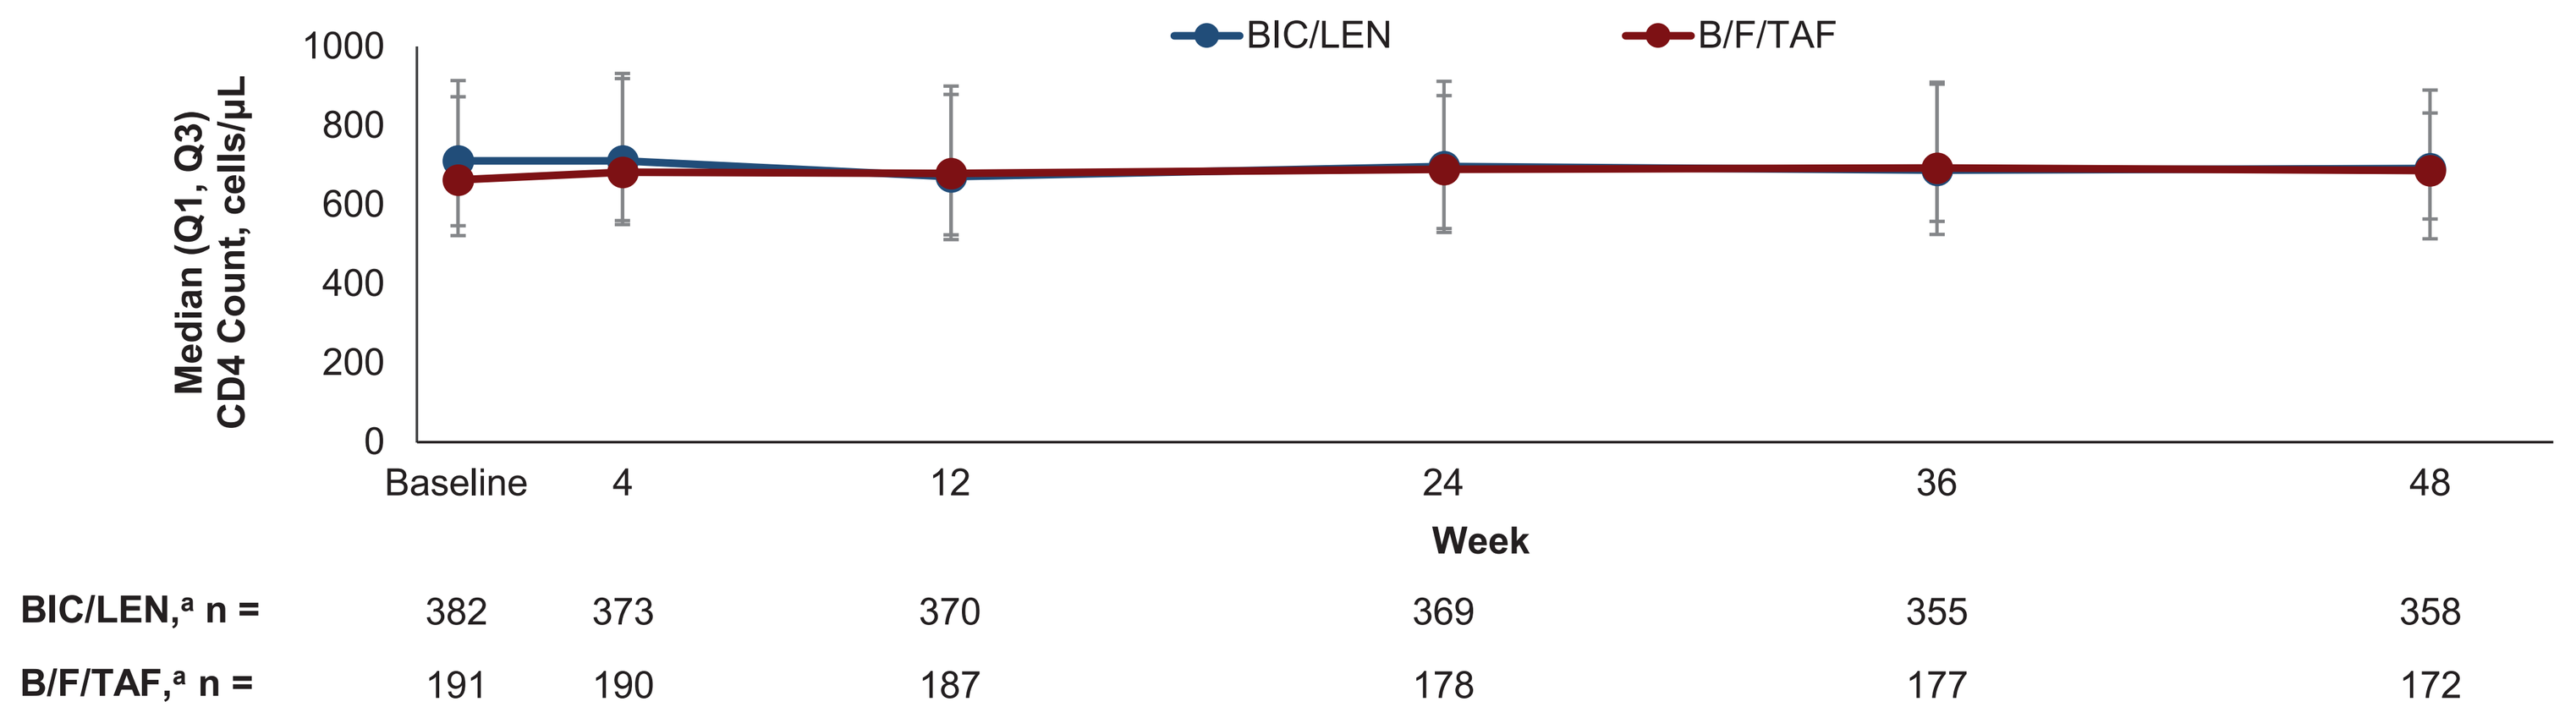
BIC/LEN
B/F/TAF
Median (Q1, Q3) CD4 Count, cells/μL
1000
800
600
400
200
0
Baseline
4
12
24
36
48
Week
BIC/LEN,[a] n = 382 373 370 369 355 358
B/F/TAF,[a] n = 191 190 187 178 177 172

[a]On-treatment values included all available data for ongoing participants or up to the last dose date plus 1 day for participants who prematurely discontinued study drug.
BIC, bictegravir; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; CD4, cluster of differentiation 4; LEN, lenacapavir; Q, quartile.

CD4 cell count remained stable in both treatment groups through 48 weeks

Post-Baseline Resistance Analyses

- Two participants on BIC/LEN and two participants on B/F/TAF underwent resistance analysis
- Of these, one participant on BIC/LEN and both participants on B/F/TAF had no treatment-emergent resistance through Week 48
- One participant on BIC/LEN had an isolated R263K integrase substitution at Week 36
 - This participant had a history of prior exposure to raltegravir and dolutegravir
 - The R263K isolate remained phenotypically susceptible to BIC
 - No capsid mutations were detected

Summary of AEs

Number (%) of participants with any:	BIC/LEN n = 383	B/F/TAF n = 191
AE	288 (75.2)	141 (73.8)
AE of Grade 3 or higher	38 (9.9)	15 (7.9)
Serious AE	27 (7.0)	13 (6.8)
Drug-related AE	40 (10.4)	23 (12.0)
Drug-related AE of Grade 3 or higher	1 (0.3)[a]	0
Serious drug-related AE	0	0
AE leading to premature study treatment discontinuation	6 (1.6)[b]	3 (1.6)[c]
Death	0	1 (0.5)[d]

[a]Grade 3 rhabdomyolysis. [b]Deemed unrelated to study treatment: anxiety (n = 1), colon cancer (n = 1), decreased libido (n = 1), encephalopathy and intracranial mass (n = 1); deemed related to study treatment: alopecia and fatigue (n = 1), rash, restless legs syndrome and trismus (n = 1). [c]Deemed unrelated to study treatment: abdominal discomfort (n = 1), road traffic accident (n = 1); deemed related to study treatment: dry mouth (n = 1). [d]Coronary artery disease, deemed unrelated to study treatment. AE, adverse event; BIC, bictegravir; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; LEN, lenacapavir.

BIC/LEN was generally well tolerated, with similar rates of AEs and drug-related AEs in both treatment groups and no serious drug-related AEs; rates of discontinuation due to AEs were low

Most Common AEs

Participants, n (%)	BIC/LEN n = 383	B/F/TAF n = 191
AE with ≥ 5% frequency in the BIC/LEN group		
Diarrhea	31 (8.1)	9 (4.7)
Nasopharyngitis	30 (7.8)	13 (6.8)
Upper respiratory tract infection	29 (7.6)	9 (4.7)
Headache	22 (5.7)	6 (3.1)
Nausea	20 (5.2)	11 (5.8)

Median (Q1, Q3) exposure was 60.6 (53.0, 69.0) weeks for the BIC/LEN group and 59.9 (52.0, 67.9) weeks for the B/F/TAF group.
AE, adverse event; BIC, bictegravir; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; LEN, lenacapavir; Q, quartile.

Absolute Change From Baseline in Body Weight and BMI at Week 48

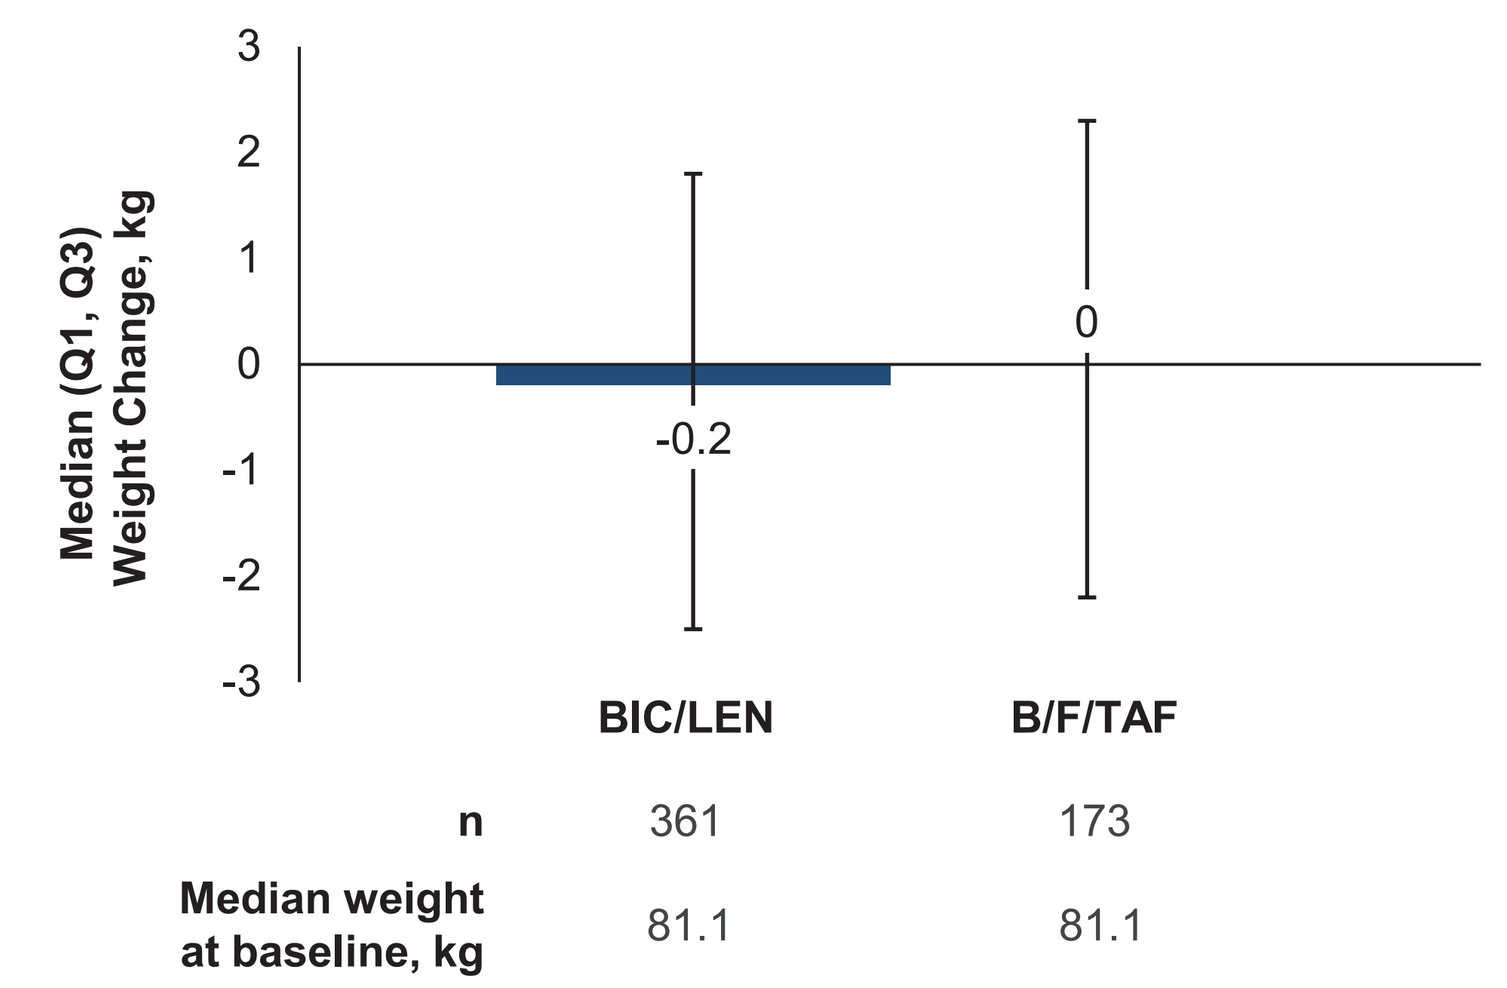
Median (Q1, Q3) Weight Change, kg
3
2
1
0
-1
-2
-3
-0.2
0
BIC/LEN
B/F/TAF
n 361 173
Median weight at baseline, kg 81.1 81.1

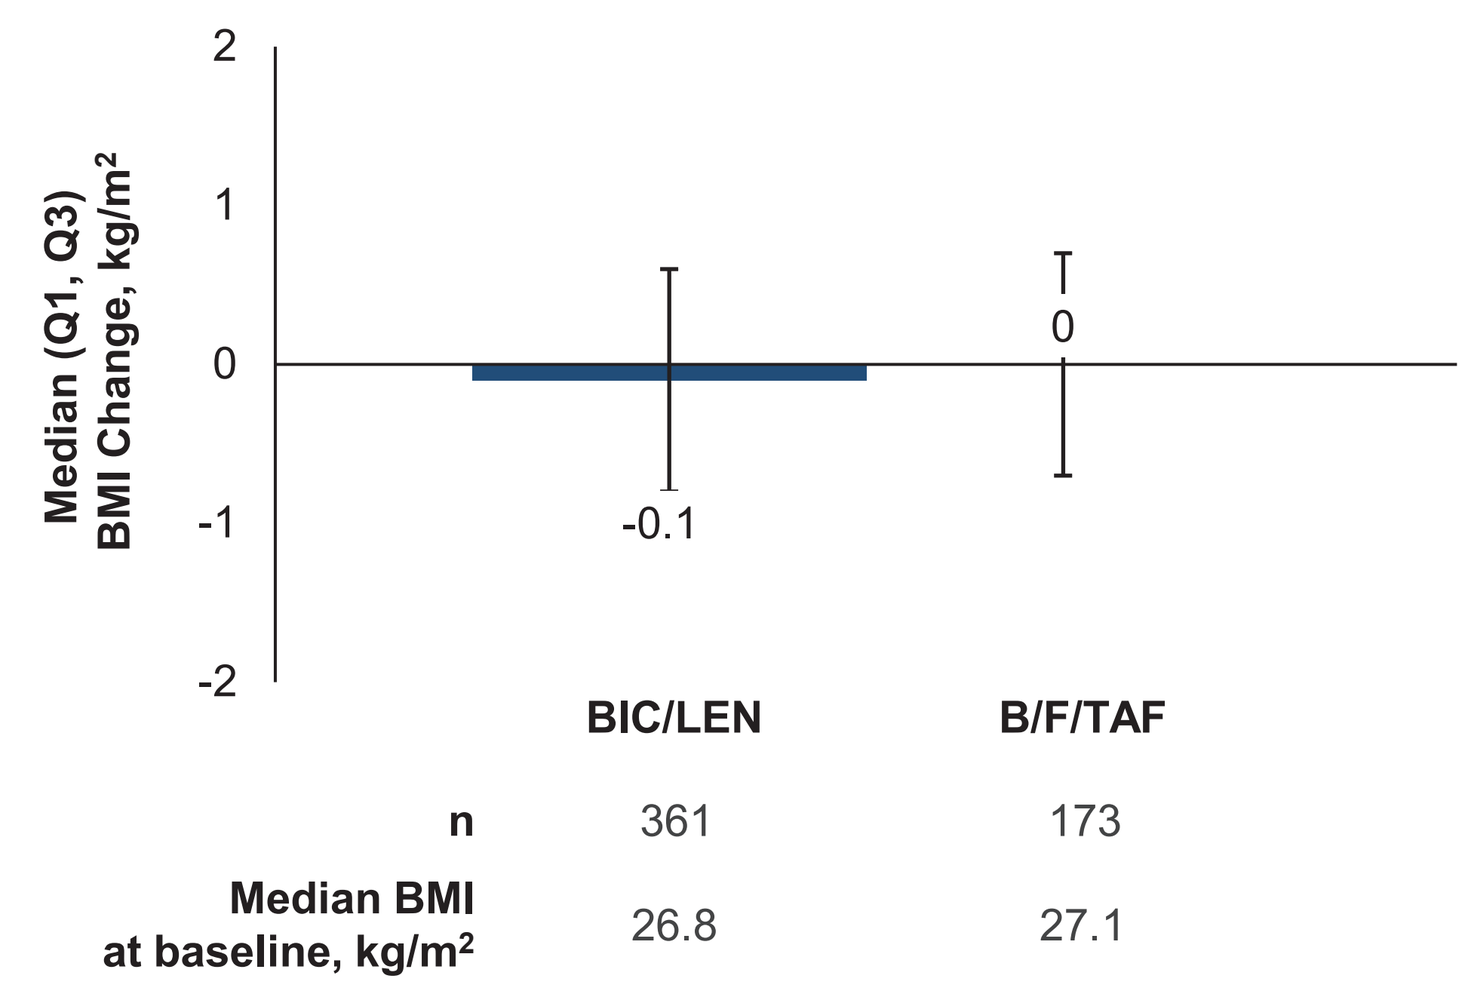
Median (Q1, Q3) BMI Change, kg/m²
2
1
0
-1
-2
-0.1
0
BIC/LEN
B/F/TAF
n 361 173
Median BMI at baseline, kg/m² 26.8 27.1

BIC, bictegravir; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; BMI, body mass index; LEN, lenacapavir; Q, quartile.

Weight and BMI remained stable in both groups through 48 weeks of treatment

References: 1. Orkin C, et al. Oral 181 presented at: CROI; Feb. 22-25, 2026; Denver, CO, USA. 2. Antinori A, et al. *Infect Dis Ther.* 2025;15:217-44. 3. Cotte L, et al. *PLOS One.* 2017;12:e0170661.

Acknowledgments: We extend our thanks to the participants, their families, and all participating investigators. We thank Mauricio Montezuma and Michelle D'Antoni of Gilead Sciences, Inc. for their contribution to the study. This study was funded by Gilead Sciences, Inc. Medical writing support was provided by Anna Chapman-Barnes, PhD, and Anne Errichelli, DPhil (Aspire Scientific Ltd, Manchester, UK), and was funded by Gilead Sciences, Inc.

Disclosures: **EGM** reports grants (paid to institution) from ViiV Healthcare and serving as the Independent Data Monitoring Committee Chair for a ViiV Healthcare–funded clinical trial. **MRa** reports consulting fees from Gilead Sciences, Inc., Shionogi, and ViiV Healthcare; and payment/honoraria for lectures, presentations, speakers' bureaus, manuscript writing, or educational events from AbbVie, Gilead Sciences, Inc., and ViiV Healthcare. **PJR** reports payment/honoraria for lectures, presentations, speakers' bureaus, manuscript writing, or educational events from Gilead Sciences, Inc. and ViiV Healthcare. **WS** has no conflicts of interest to report. **J-PR** reports grants/contracts from AbbVie; consulting fees, payment/honoraria for lectures, presentations, speakers' bureaus, manuscript writing, educational events, or expert testimony from, and participating on a data safety monitoring or advisory board

Disclosures (cont.)**:** for, Gilead Sciences, Inc., Merck, and ViiV Healthcare; support for attending meetings and/or travel from Gilead Sciences, Inc. and ViiV Healthcare; and equipment, materials, drugs, medical writing, gifts, or other services from Merck. **MO'R** reports payment/honoraria for lectures, presentations, speakers' bureaus, manuscript writing, or educational events from, and participating on a data safety monitoring or advisory board for, Gilead Sciences, Inc.; support for attending meetings and/or travel from Gilead Sciences, Inc., Janssen, and ViiV Healthcare; and serving as an unpaid member of the ASHM Board and Australian ARV Guidelines Committee. **SL** reports grants/contracts from GSK and Merck; consulting fees from GSK; and payment/honoraria for lectures, presentations, speakers' bureaus, manuscript writing, or educational events from Gador and GSK.

Disclosures (cont.)**:** **SvS** reports consulting fees for participation on advisory boards from AbbVie, Gilead Sciences, Inc., Janssen-Cilag, MSD, Novavax, Pfizer, Sanofi, Theratechnologies, and ViiV Healthcare; honoraria for presentations and lectures from AbbVie, Gilead Sciences, Inc., Janssen-Cilag, MSD, Pfizer, Sanofi, Theratechnologies, and ViiV Healthcare; and support for attending meetings from Gilead Sciences, Inc. **KA**, **HH**, **KM**, **PS**, and **MRh** are employees of, and own stocks in, Gilead Sciences, Inc. **C-CH** reports grants (paid to institution) and consulting fees/honoraria for lectures, speakers' bureaus, or educational events from Gilead Sciences, Inc.

Correspondence: Eric G Meissner, meissner@musc.edu.

American Conference for the Treatment of HIV (ACTHIV), April 30 - May 2, 2026, Atlanta, GA, USA